532P

Analyses of Patient-Reported Outcomes (PROs) With Mirvetuximab Soravtansine (MIRV) Versus Standard Chemotherapy in the Randomized Phase 3 FORWARD I Study in Ovarian Cancer (GOG 3011)

Kathleen N. Moore,[1] Amit M. Oza,[2] Nicoletta Colombo,[3] Ana Oaknin,[4] Giovanni Scambia,[5] Domenica Lorusso,[6] Susana Banerjee,[7] Conleth G. Murphy,[8] Jason A. Konner,[9] Peter C. Lim,[10] Monica Prasad-Hayes,[11] Bradley J. Monk,[12] Jiuzhou Wang,[13] Michael J. Birrer,[14] Ignace Vergote[15]

[1]College of Medicine, The University of Oklahoma, Oklahoma City, OK, USA; [2]University Health Network, Princess Margaret Cancer Centre, Toronto, Ontario, Canada; [3]University of Milan-Bicocca, Milan, Italy; [4]Gynaecologic Cancer Programme, Vall d'Hebron Institute of Oncology (VHIO), Hospital Universitari Vall d'Hebron, Vall d'Hebron Barcelona Hospital Campus, Barcelona, Spain; [5]Agostino Gemelli University Policlinic, Rome, Italy; [6]Fondazione Policlinico Gemelli IRCCS, Rome, Italy; [7]Royal Marsden Hospital, London, UK; [8]Bon Secours Cork and Cancer Trials Ireland; [9]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [10]Center Of Hope, Reno, NV, USA; [11]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [12]University of Arizona College of Medicine, Phoenix, AZ, USA; [13]ImmunoGen, Inc., Waltham, MA, USA; [14]Winthrop P. Rockefeller Cancer Institute, University of Arkansas Medical School, Little Rock, AR, USA; [15]Katholieke Universiteit Leuven, Leuven, Belgium

BACKGROUND

- Mirvetuximab soravtansine (MIRV) is a first-in-class antibody-drug conjugate (ADC) comprising a folate receptor alpha (FRα)-binding antibody, cleavable linker, and maytansinoid DM4 payload, a potent tubulin-targeting agent[1]
- MIRV has demonstrated clinically meaningful antitumor activity with a favorable safety profile in patients with FRα-positive[a] ovarian cancer as monotherapy and in combination[2,3]
- During the phase 3 FORWARD I trial, patient-reported outcomes (PROs) regarding chemotherapy side effects, cancer-specific symptoms, and quality of life (QoL) were collected[4]
- Preplanned and exploratory analyses of PRO data were conducted to determine differences with MIRV vs chemotherapy and to inform the collection of PRO data in subsequent trials with MIRV (eg, phase 3 MIRASOL; NCT04209855)

[a]Antitumor activity with MIRV has been demonstrated with single-agent MIRV in FRα-high PROC (≥75% tumor cells FRα-positive by PS2+)[2] and in combination with other agents in FRα low-to-high PROC (≥25% tumor cells FRα-positive by PS2+).[3]

Methods

- The phase 3, open-label, randomized trial FORWARD I (N=366; NCT02631876) enrolled patients with platinum-resistant FRα-positive advanced epithelial ovarian cancer (EOC)[a]
 - Patients were randomized 2:1 to receive MIRV (n=248; 6 mg/kg AIBW[b] once every 3 weeks) or investigator's choice of chemotherapy (IC chemo[c], n=118)
- Patients completed PRO assessments (Table 1) during screening, on day 1 of cycle 1, every 9 weeks thereafter (±1 week) until disease progression, and at the end of treatment visit
- Differences between treatment groups (LPP analysis population[d]) were assessed by descriptive statistics, Chi-squared analysis, Fisher's exact test, stratified Kaplan-Meier curves, unadjusted log-rank *P* values, and odds ratios

[a]In addition to EOC, patients with primary peritoneal cancer and/or fallopian tube cancer were also considered for enrollment. [b]AIBW, also known as AdjBW, is calculated as IBW (kg) + 0.4 (actual weight - IBW). IBW for females is calculated as 0.9*height (cm) - 92. [c]Investigator's choice of chemotherapy was specified by investigators before randomization: paclitaxel, 80 mg/m² on days 1, 8, and 15 every 4 weeks; PLD, 40 mg/m² once every 4 weeks; topotecan, 4 mg/m² on days 1, 8, and 15 every 4 weeks or 1.25 mg/m² on days 1 to 5 every 3 weeks. [d]The LPP analysis included all patients within each of the parent populations (ITT and FRα high) who (1) survived, (2) remained in the study from screening through to the day of a given assessment, and (3) had available QoL data for baseline and day of assessment.

Table 1. FORWARD I PRO Assessments

PRO assessment	Description
EORTC QLQ-C30 (C30)	A 30-item questionnaire designed to assess the QoL in patients with cancer by measuring functional domains, symptoms, and global QoL/health status
EORTC QLQ-OV28 (OV28)	A 28-item ovarian cancer supplemental module developed to augment the C30 with 3 multi-item functional scales and 5 multi-item symptom scales
FOSI	An 8-item measure of symptom response to treatment for ovarian cancer

PRO Analyses

Primary: Minimally important difference (MID) response in abdominal/GI symptoms at week 8/9 by OV28 Abdominal/GI symptom subscale score:
- ≥15-point increase: Improved
- <15-point increase: Not improved

Secondary: Time to symptom worsening

Patient Demographics and Baseline Characteristics

Table 2. Baseline Clinical Characteristics

Characteristic	MIRV (n=248)	IC chemo (n=118)
Age, median (range), y	64 (34-89)	64 (31-86)
Primary cancer diagnosis, n (%)		
Epithelial ovarian cancer	207 (83.5)	105 (89.0)
Fallopian tube cancer	14 (5.6)	5 (4.2)
Primary peritoneal cancer	27 (10.9)	8 (6.8)
ECOG PS, n (%)[a]		
0	141 (56.9)	60 (50.8)
1	106 (42.7)	57 (48.3)
No. of prior systemic therapies, n (%)[b]		
1 or 2	159 (64.1)	74 (62.7)
3	86 (34.7)	43 (36.4)
FRα expression by 10× scoring, n (%)[c]		
High	147 (59.3)	71 (60.2)
Medium	101 (40.7)	46 (39.0)
Prior exposure, n (%)		
Platinum	248 (100)	118 (100)
Paclitaxel	238 (96.0)	113 (95.8)
Bevacizumab	121 (48.8)	55 (46.6)
PARPi	44 (17.7)	19 (16.1)

[a]Performance status data not available for one patient in each arm. [b]Four patients enrolled were ineligible due to >3 prior lines of therapy. [c]Medium, 50% to 74%; high ≥75% of tumor cells with any FRα membrane staining visible at ≤10× microscope objective; one patient randomized to the IC chemo arm was subsequently determined to have a FRα expression level <50%.

Results: OV28 Abdominal/GI Scale

- The proportion of patients with a ≥15-point improvement on the OV28 Abdominal/GI scale at week 8/9 was significantly higher in the MIRV ITT group vs IC chemo (Figure 1)
- Patients with high FRα expression who were treated with MIRV demonstrated similar improvements; however, these findings did not reach statistical significance compared with IC chemo (Figure 1)

Figure 1. Improvement in the OV28 Abdominal/GI Symptom Subscale by Treatment Group at Week 8/9

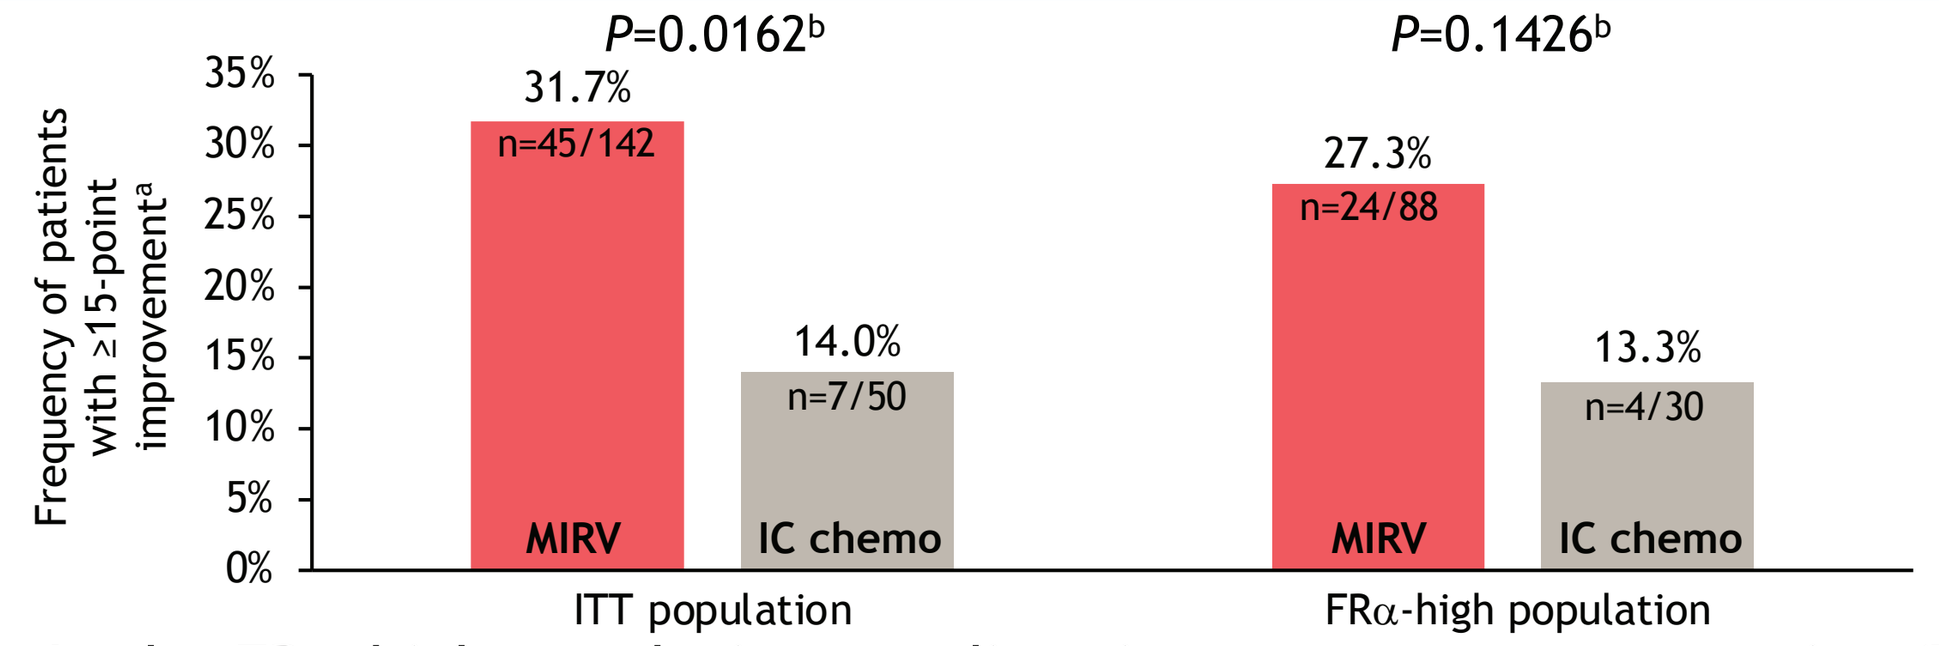
P=0.0162[b]
P=0.1426[b]
Frequency of patients with ≥15-point improvement[a]
35%
30%
25%
20%
15%
10%
5%
0%
31.7%
n=45/142
MIRV
14.0%
n=7/50
IC chemo
27.3%
n=24/88
MIRV
13.3%
n=4/30
IC chemo
ITT population
FRα-high population

[a]Improvement was defined as ≥15-point increase from baseline. Any change <15 points was categorized as unimproved. [b]*P* value from chi-square test or Fisher exact test if necessary.

- In the FRα-high population, median time to symptom worsening (TSW) on the OV28 Abdominal/GI symptom subscale was nearly 2 months longer with MIRV compared with IC chemo (Figure 2b), but there was no significant difference between treatment groups in the ITT population (Figure 2a)

Figure 2a. ITT Population: TSW on the OV28 Abdominal/GI Symptom Subscale

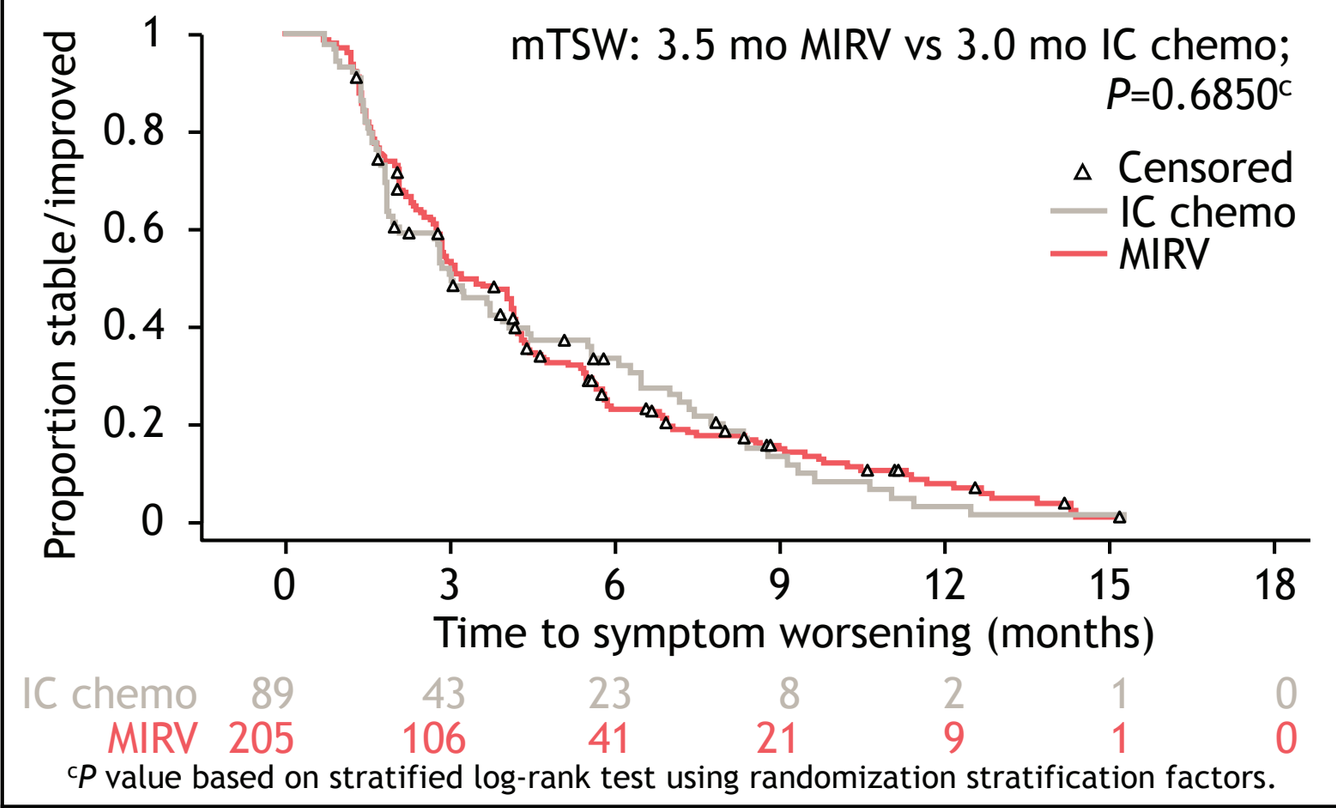
mTSW: 3.5 mo MIRV vs 3.0 mo IC chemo;
P=0.6850[c]
Censored
IC chemo
MIRV
Proportion stable/improved
Time to symptom worsening (months)
0 3 6 9 12 15 18
IC chemo 89 43 23 8 2 1 0
MIRV 205 106 41 21 9 1 0

Figure 2b. FRα-High Population: TSW on the OV28 Abdominal/GI Symptom Subscale

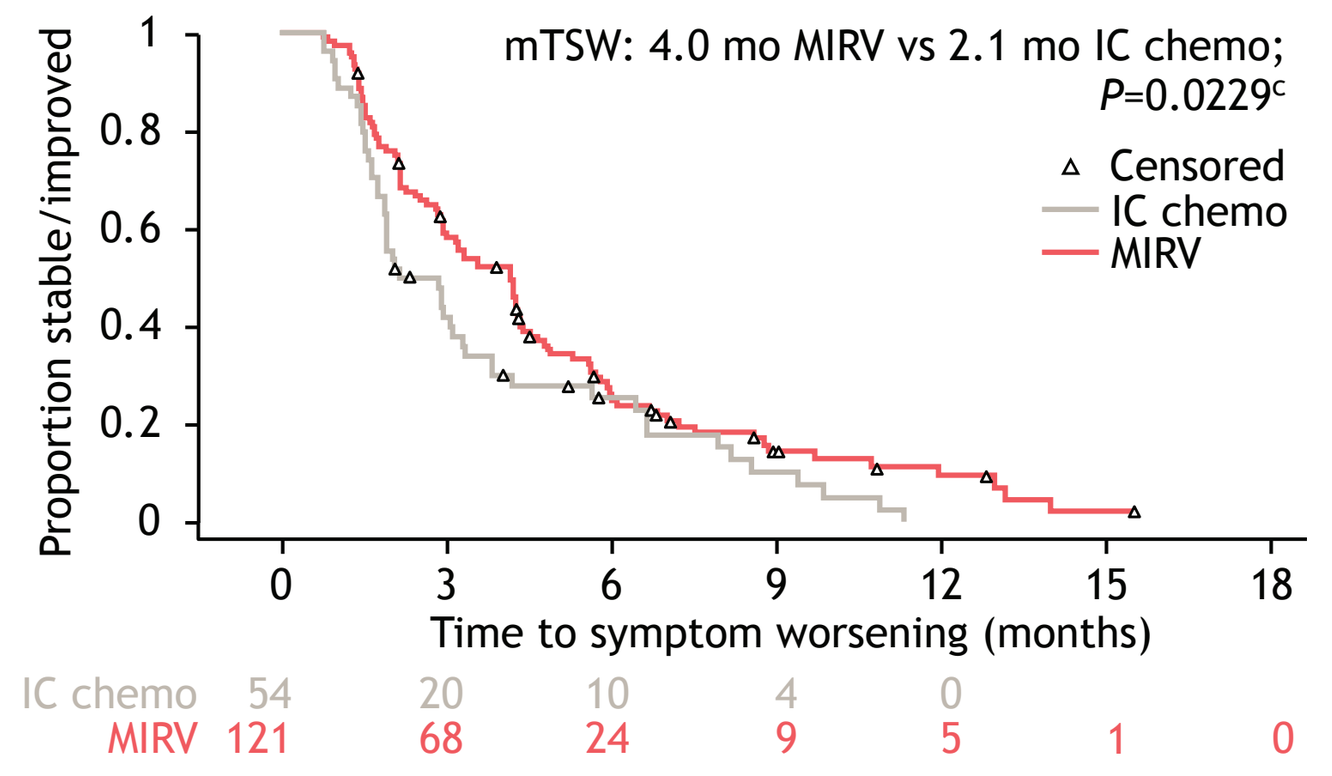
mTSW: 4.0 mo MIRV vs 2.1 mo IC chemo;
P=0.0229[c]
Censored
IC chemo
MIRV
Proportion stable/improved
Time to symptom worsening (months)
0 3 6 9 12 15 18
IC chemo 54 20 10 4 0
MIRV 121 68 24 9 5 1 0

[c]*P* value based on stratified log-rank test using randomization stratification factors.

Results: Likelihood of Symptom Deterioration

- Compared with the respective IC chemo populations, the likelihood of deterioration of abdominal/GI symptoms on the OV28 was (Figure 3):
 - 70% lower in the MIRV ITT population (95% CI, 0.15-0.60; *P*=0.0007)
 - 80% lower in the MIRV FRα-high population (95% CI, 0.10-0.54; *P*=0.0007)
- Similar results for the likelihood of symptom deterioration with MIRV were observed on all other OV28 subscales, including chemotherapy side effects, sexuality, hair loss, pain severity (peripheral neuropathy), and body image (Figure 3)
- General improvements were also observed with MIRV on the ovarian cancer-specific FOSI assessment (Figure 3)

Figure 3. Odds Ratios for Categorical Change on the OV28 and FOSI: MIRV vs IC Chemo in the Longitudinal Period Population

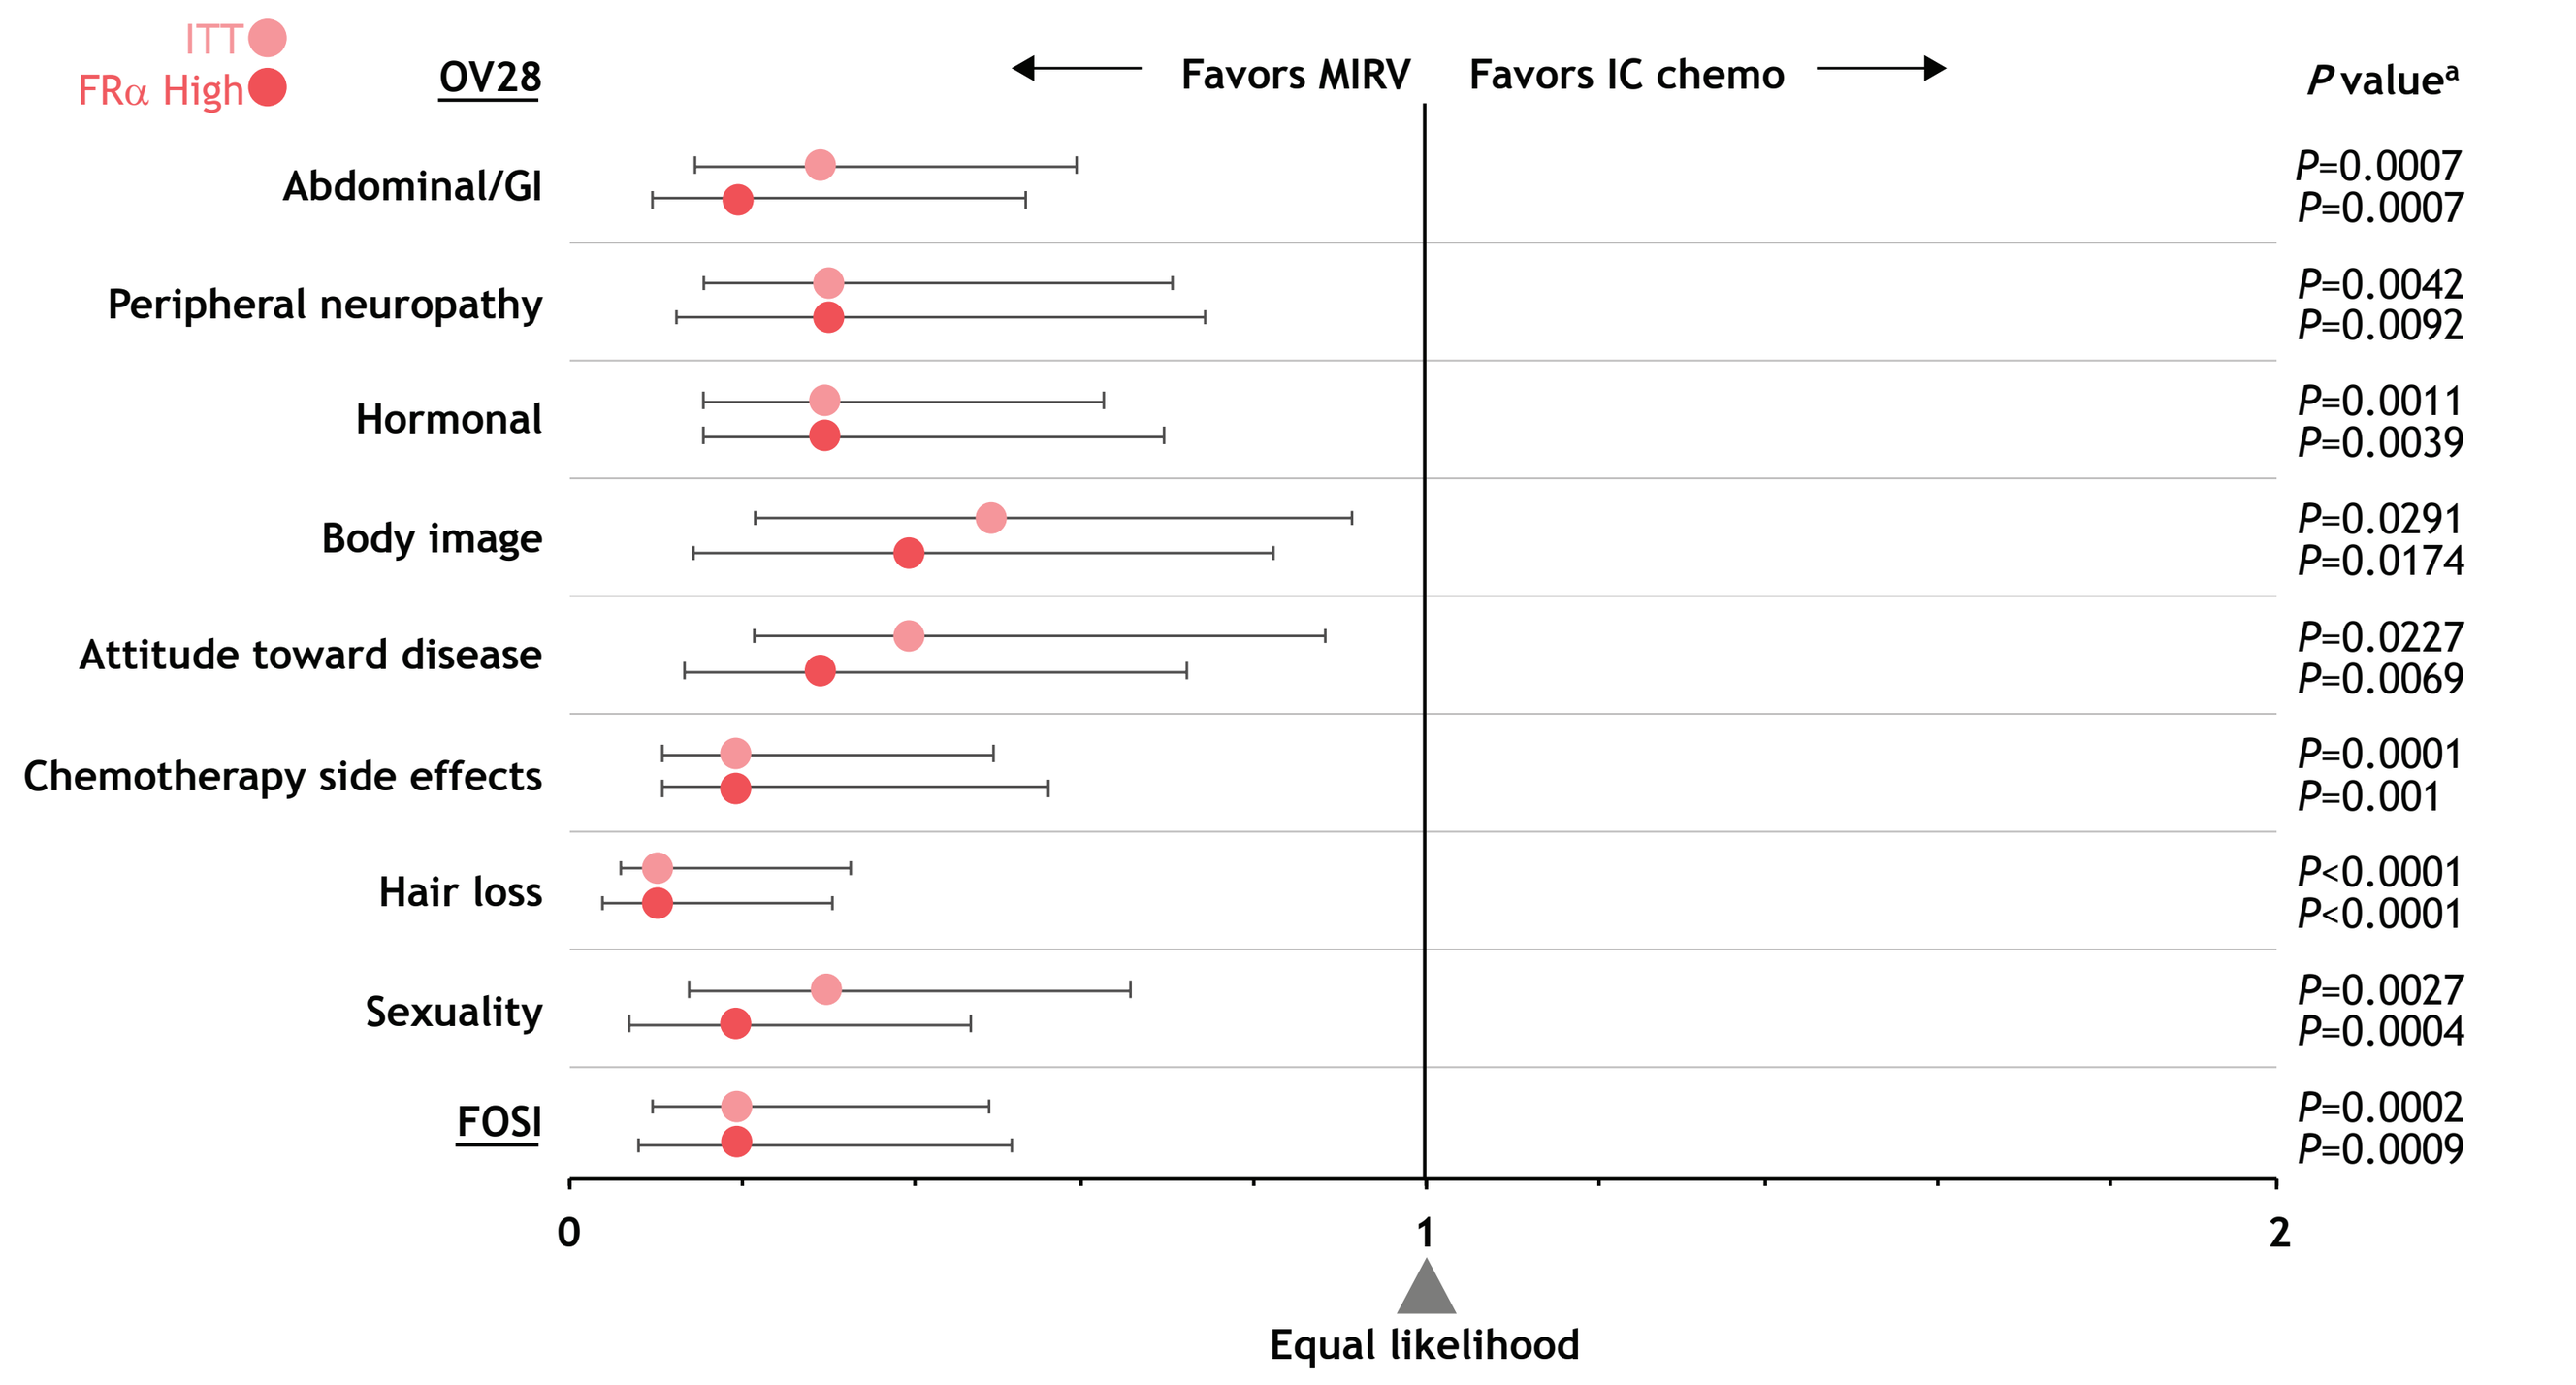
ITT
FRα High
OV28
Favors MIRV
Favors IC chemo
P value[a]
Abdominal/GI
P=0.0007
P=0.0007
Peripheral neuropathy
P=0.0042
P=0.0092
Hormonal
P=0.0011
P=0.0039
Body image
P=0.0291
P=0.0174
Attitude toward disease
P=0.0227
P=0.0069
Chemotherapy side effects
P=0.0001
P=0.001
Hair loss
P<0.0001
P<0.0001
Sexuality
P=0.0027
P=0.0004
FOSI
P=0.0002
P=0.0009
0
1
2
Equal likelihood

[a]*P* value for Wald chi-square test associated with mixed model parameter estimate β(treatment)= 0.

Results: Changes in C30 Physical Functioning Scores

- Analyses of C30 demonstrated a statistically significant benefit in physical functioning for MIRV over IC chemo in both the ITT and FRα-high populations

Table 3. Model-Based Change from Baseline on C30 Physical Functioning Scale by Treatment Group Over All Cycles[a]

C30 physical functioning (revised)	ITT population		FRα-high population	
	MIRV (n=248)	IC chemo (n=118)	MIRV (n=147)	IC chemo (n=71)
LS mean overall change (SE)	-0.21 (2.59)	-4.31 (2.89)	-0.79 (4.71)	-6.81 (5.11)
LS mean difference from IC chemo (SE)	4.10 (1.96)		6.03 (2.33)	
95% CI	0.25-7.96		1.42-10.63	
P value[b]	0.0369		0.0106	

[a]The primary model includes treatment, time (continuous), treatment*time, baseline PRO score, age, race, and 3 stratification variables (prior lines of therapy, FRα levels, and IC chemo). [b]*P* value for LS mean treatment difference = 0.

Results: Categorical Changes and TSW on the FOSI

- Categorical change analyses of FOSI scores demonstrated that by cycle 7:
 - 88.9% of ITT population patients on IC chemo had declined vs 70.3% with MIRV
 - 88.1% of FRα-high population patients on IC chemo had declined vs 65.0% with MIRV

Figure 4a. ITT Population: TSW on the FOSI

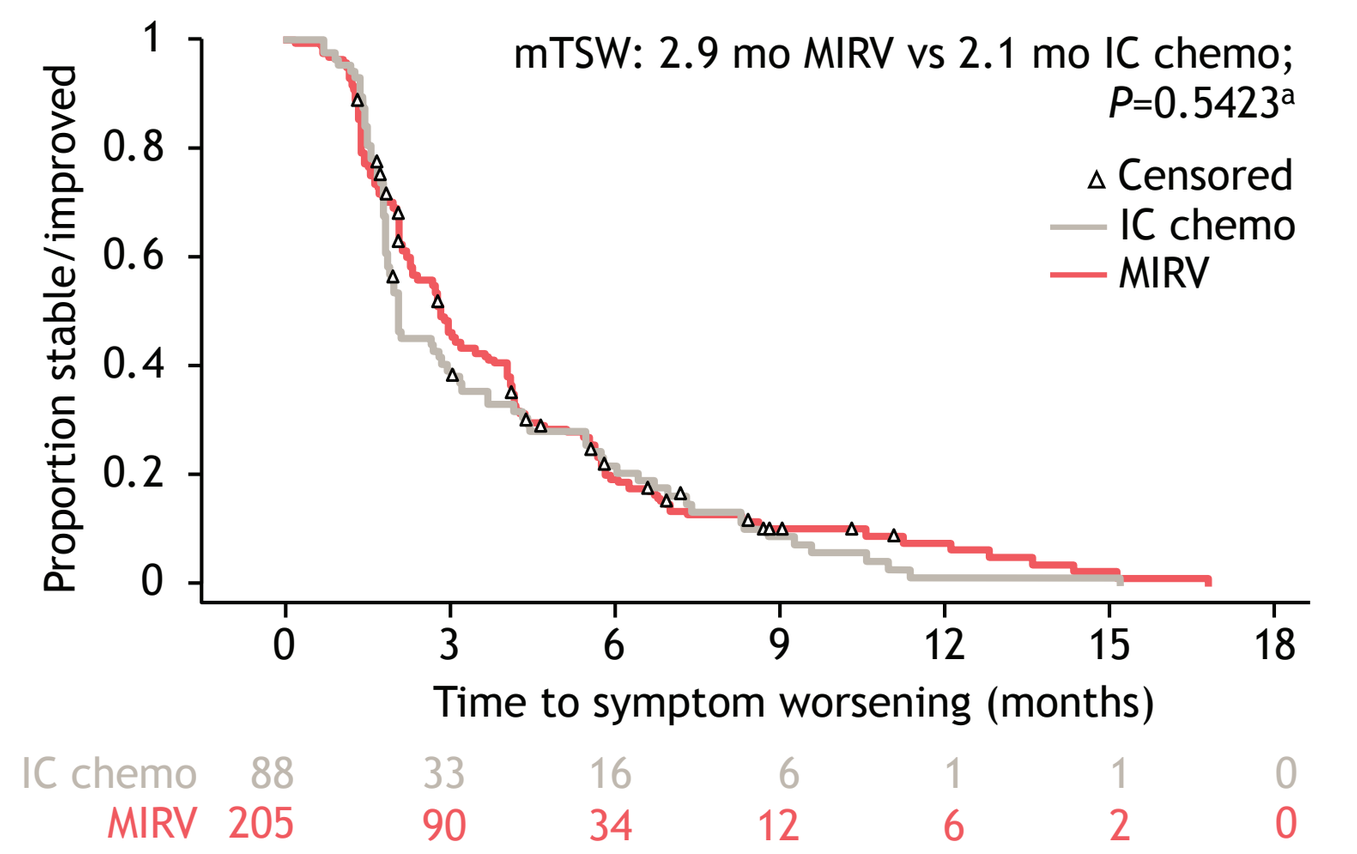
mTSW: 2.9 mo MIRV vs 2.1 mo IC chemo;
P=0.5423[a]
Censored
IC chemo
MIRV
Proportion stable/improved
Time to symptom worsening (months)
0 3 6 9 12 15 18
IC chemo 88 33 16 6 1 1 0
MIRV 205 90 34 12 6 2 0

Figure 4b. FRα-High Population: TSW on the FOSI

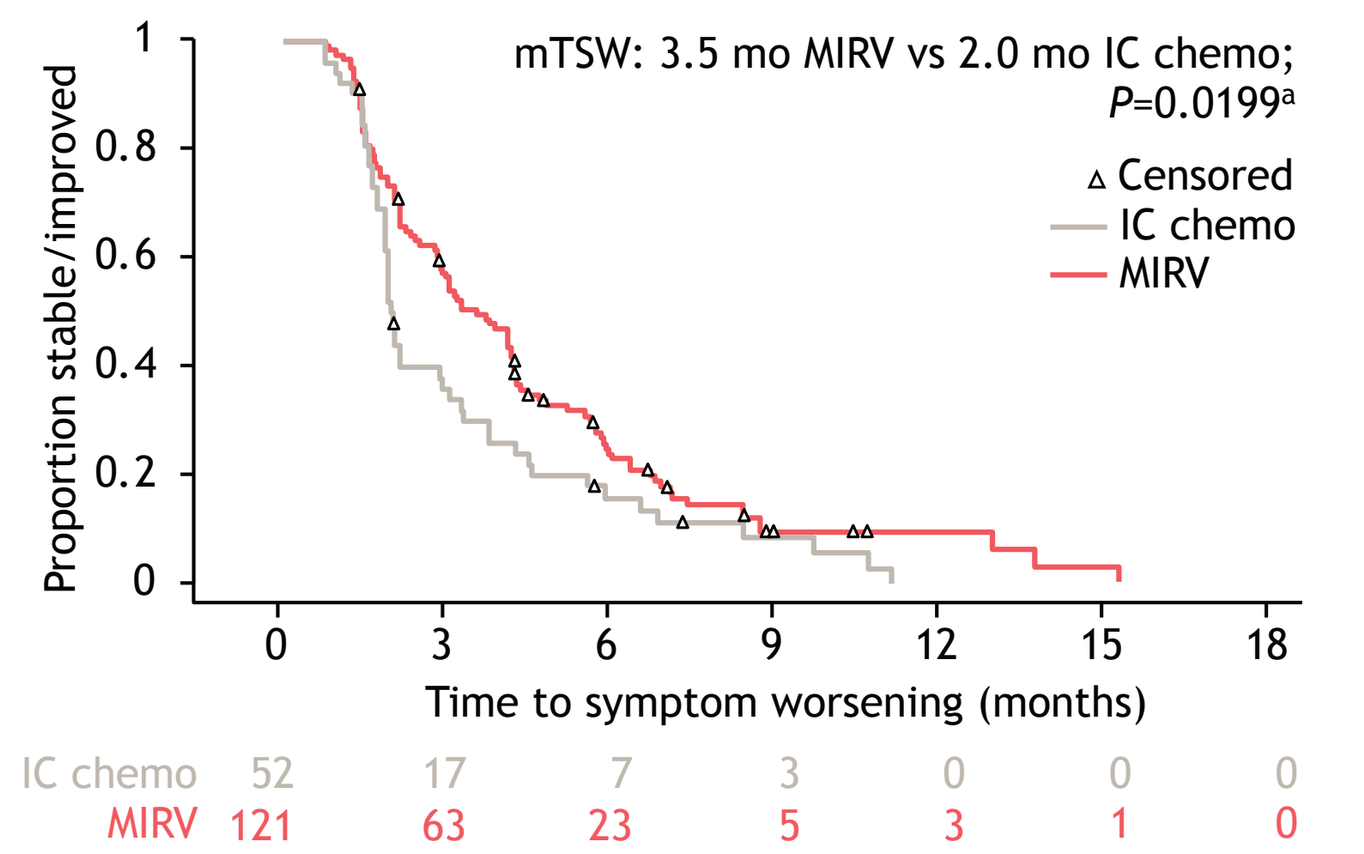
mTSW: 3.5 mo MIRV vs 2.0 mo IC chemo;
P=0.0199[a]
Censored
IC chemo
MIRV
Proportion stable/improved
Time to symptom worsening (months)
0 3 6 9 12 15 18
IC chemo 52 17 7 3 0 0 0
MIRV 121 63 23 5 3 1 0

[a]*P* value based on stratified log-rank test using randomization stratification factors.

CONCLUSIONS

- Analysis of PROs from FORWARD I found that MIRV demonstrated a statistically significant benefit over IC chemo for the number of patients achieving a 15-point improvement on the OV28 Abdominal/GI symptom subscale at week 8/9
- Improved PROs were observed with MIRV treatment compared with IC chemo across multiple symptoms, including chemotherapy side effects, sexuality, hair loss, pain severity, body image, and general improvement in ovarian cancer–specific symptoms on the FOSI
- Symptom benefits occurring with MIRV, such as time to symptom worsening, were observed exclusively or more profoundly in the FRα-high population
- The results from these analyses will inform the design of future PRO analyses with MIRV

These positive PRO findings, in conjunction with the safety profile of MIRV in recurrent ovarian cancer along with significant antitumor activity support MIRV as a potential new standard of care for patients with FRα-positive ovarian cancer

Abbreviations: ADC, antibody-drug conjugate; AdjBW, adjusted ideal body weight; AIBW, adjusted ideal body weight; C30, EORTC Quality of Life Questionnaire-core 30; DM4, N2'-[4-[(3-carboxypropyl)dithio]-4-methyl-1-oxo-2-sulfopentyl]-N2'-deacetylmaytansine; ECOG PS, Eastern Cooperative Oncology Group Performance Status; EOC, epithelial ovarian cancer; EORTC, European Organisation for Research and Treatment of Cancer; FOSI, Functional Assessment of Cancer Therapy – Ovarian Symptom Index; FRα, folate receptor alpha; GI, gastrointestinal; IBW, ideal body weight; IC chemo, investigator's choice of chemotherapy; ITT, intention to treat; LPP, Longitudinal Period Population; LS, least squares; MID, minimally important difference; MIRV, mirvetuximab soravtansine; mTSW, median time to symptom worsening; OV28, EORTC Quality of Life Questionnaire-Ovarian Cancer Module; PARPi, poly (adenosine diphosphate [ADP]-ribose) polymerase inhibitor; PLD, pegylated liposomal doxorubicin; PRO, patient-reported outcome; PS2+, positive staining intensity ≥2; QoL, quality of life; TSW, time to symptom worsening.

References: 1. Moore KN, et al. *Cancer.* 2017;123(16):3080-3087. 2. Matulonis UA, et al. Presented at: 2022 American Society of Clinical Oncology Annual Meeting; June 3-7, 2022; Chicago, IL. Poster 391. 3. Matulonis UA, et al. Presented at: 2018 European Society for Medical Oncology Congress; October 19-23, 2018; Munich, Germany. Abstract 949P. 4. Moore KN, et al. *Ann Oncol.* 2021;32(6):757-765.

Dr Moore reports the following financial relationships: Consulting/Advising with Aravive, Alkermes, AstraZeneca, Blueprint Pharma, Clovis, Eisai, Elevar, GSK/Tesaro, Genentech/Roche, Hengrui, ImmunoGen, InxMed, Imab, Lilly, Mereo, Merck, Myriad, Mersana, OncXerna, Oncomed, OncoNova, Novartis, Tarveda, VBL Therapeutics, and Verastem; Research funding from PTC Therapeutics, Lilly, GSK/Tesaro, and Merck; Associate directorship with GOG Partners; Sits on director's board for The GOG Foundation, Inc.

Acknowledgements: The study described here was sponsored by ImmunoGen, Inc. The authors would like to especially thank the patients who consented to be included in the FORWARD I trial as well as their families. Editorial assistance in the preparation of this poster was provided by PRECISIONscientia, funded by ImmunoGen, Inc. Studies described here were sponsored by ImmunoGen, Inc. Copies of this poster (including those obtained through Quick Response [QR] Code) are for personal use only and may not be reproduced without permission from ESMO or the author of this poster.

ESMO Congress September 9-13, 2022

©2022 ImmunoGen, Inc., Waltham, MA, USA