Poster
316

Treatment of Tumor Cells with Mirvetuximab Soravtansine, a FRα-Targeting Antibody-Drug Conjugate (ADC), Activates Monocytes Through Fc-FcγR Interaction and Immunogenic Cell Death

Anna Skaletskaya, Jose Ponte, Thomas Chittenden, Yulius Setiady

IMMUNOGEN

INTRODUCTION

- Mirvetuximab soravtansine (IMGN853) is an ADC comprising the humanized FRα-binding M9346A antibody linked to the tubulin-disrupting maytansinoid, DM4.
- IMGN853 binds to FRα on cancer cells and is internalized; DM4 is released through enzymatic degradation of the antibody and linker cleavage, resulting in disruption of cell division and cell death.
- IMGN853 showed encouraging single-agent activity and was generally well-tolerated in FRα-positive ovarian cancer patients in a Phase 1 study (NCT01609556).
- IMGN853 is entering FORWARD I, a Phase 3 monotherapy study, and is also being evaluated in combination with other agents, including the checkpoint inhibitor pembrolizumab, in a Phase 1b/2 study, FORWARD II (NCT02606305).
- This study explored potential mechanism(s) whereby IMGN853 can show enhanced activity in combination with a checkpoint inhibitor.

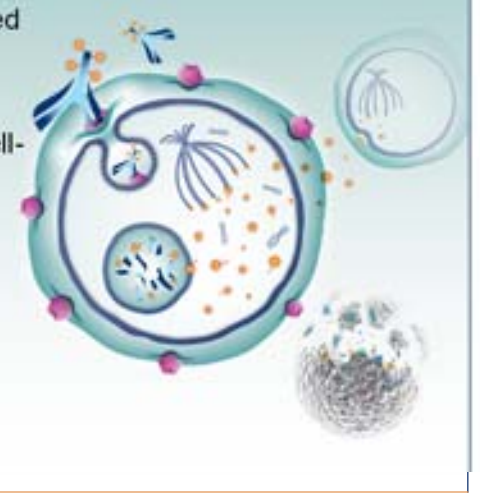

IMGN853 Treatment of KB Cells Activates Peripheral Blood Monocytes

- Peripheral blood mononuclear cells (PBMC) from normal donors were incubated with FRα-positive KB cells at 30:1 ratio in the presence or absence of test articles for 48 hours, stained for various phenotypic and activation markers, and analyzed by flow cytometry.
- CD14+ monocytes showed a significant difference in activation measured by CD86 level.

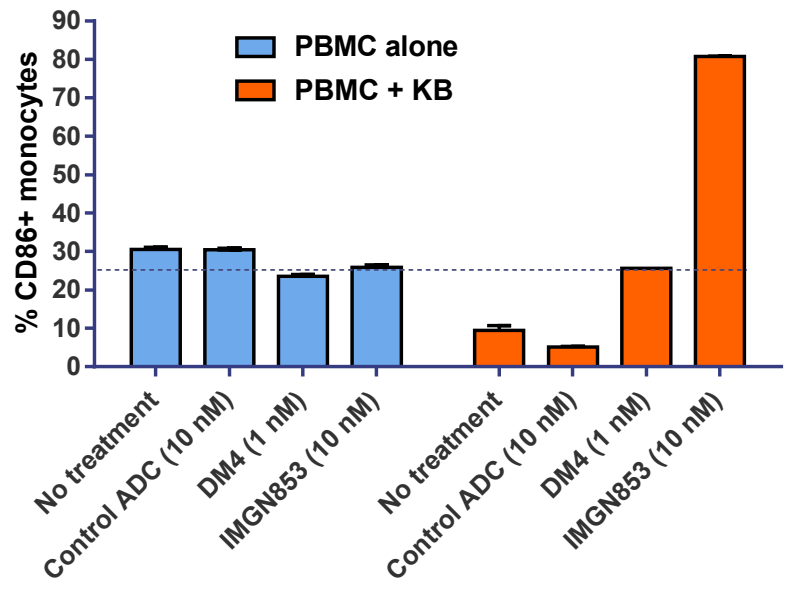
% CD86+ monocytes
PBMC alone
PBMC + KB
No treatment
Control ADC (10 nM)
DM4 (1 nM)
IMGN853 (10 nM)

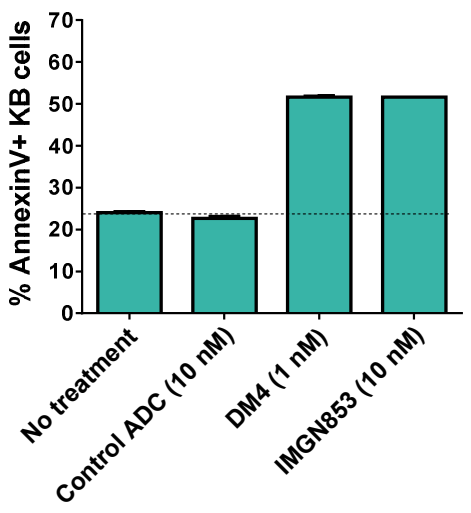
% AnnexinV+ KB cells
No treatment
Control ADC (10 nM)
DM4 (1 nM)
IMGN853 (10 nM)

IMGN853 Treatment of KB Cells Directly Activates Monocytes

Monocytes were isolated from peripheral blood of normal donors by negative selection and incubated with KB cells at a 3:1 ratio in the presence of IMGN853 (10 nM) for 48 hours, stained for CD14 and CD86, and analyzed by flow cytometry

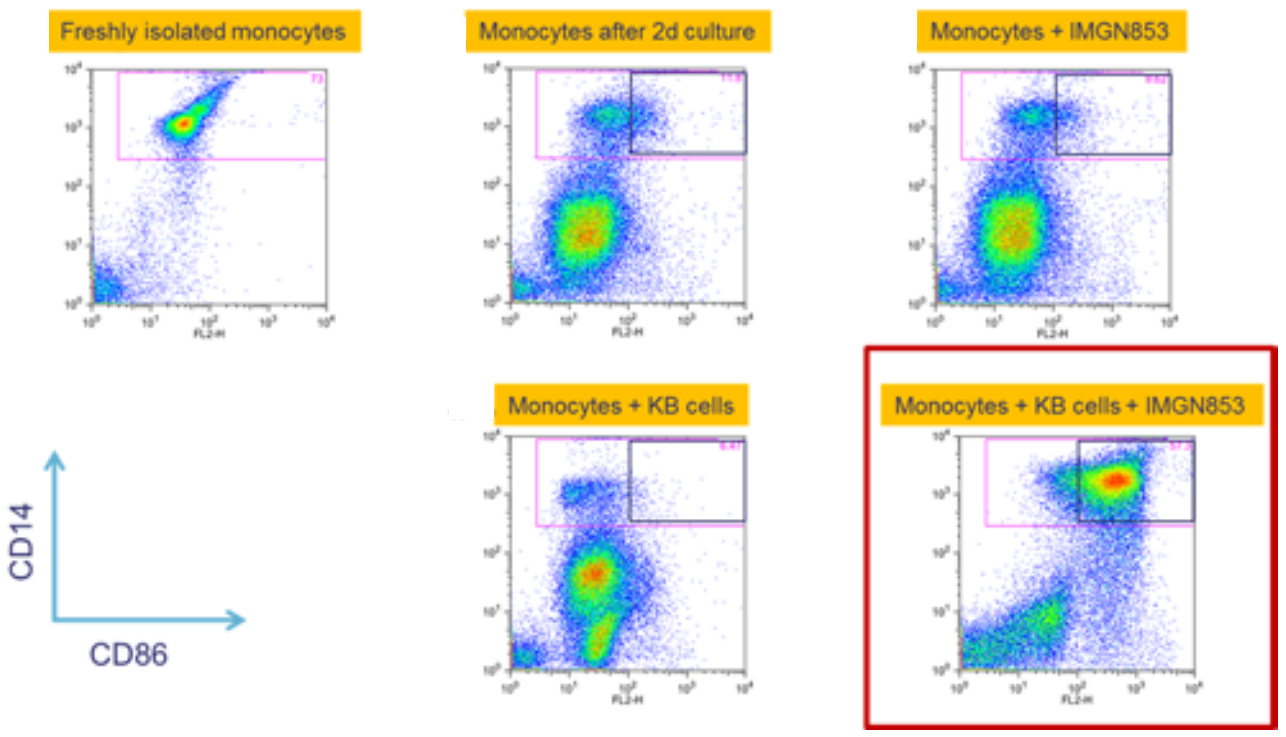
Freshly isolated monocytes
Monocytes after 2d culture
Monocytes + IMGN853
Monocytes + KB cells
Monocytes + KB cells + IMGN853
CD14
CD86

Monocyte Activation Depends on M Ab's Fc and DM4

Monocytes were isolated from peripheral blood of normal donors by negative selection and incubated with KB cells at a 3:1 ratio in the presence of IMGN853 (10 nM) or M9346A (M) Ab (10 nM) or free maytansinoid (DM4) (1 nM) for 48 hours and stained for CD14 and CD86.

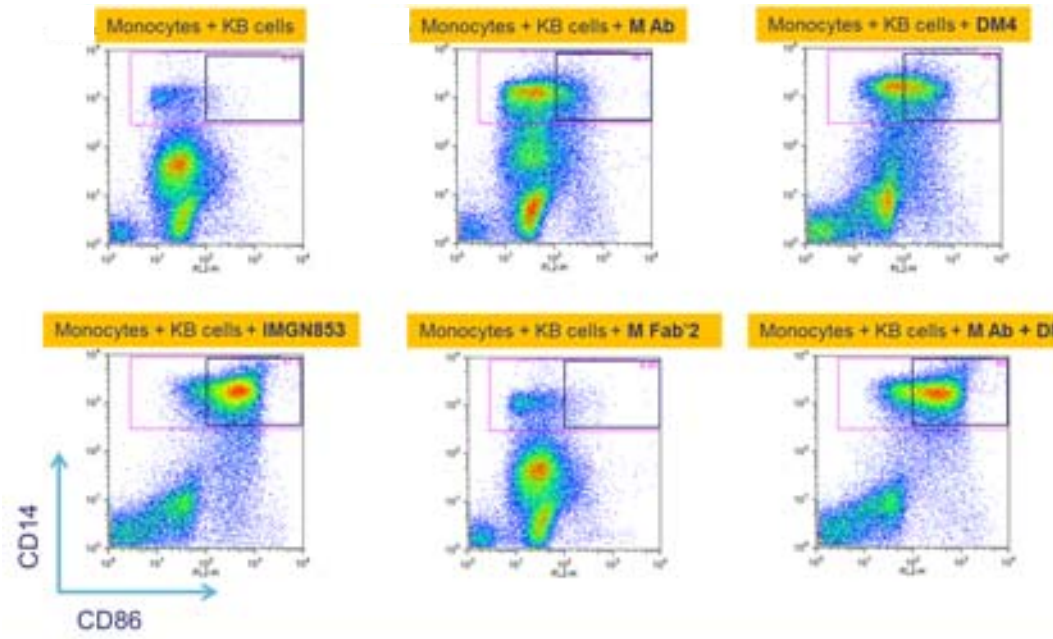
Monocytes + KB cells
Monocytes + KB cells + M Ab
Monocytes + KB cells + DM4
Monocytes + KB cells + IMGN853
Monocytes + KB cells + M Fab'2
Monocytes + KB cells + M Ab + DM4
CD14
CD86

Phagocytosis of KB Cells by Monocytes Depends on Fc-FcγR interaction and DM4

Monocytes were isolated from peripheral blood of normal donors by negative selection and incubated with PKH26-labelled KB cells at a 3:1 ratio in the presence of IMGN853 (10 nM) or M9346A (M) Ab (10 nM) or free maytansinoid (DM4) (1 nM) for 48 hours and stained for CD14.

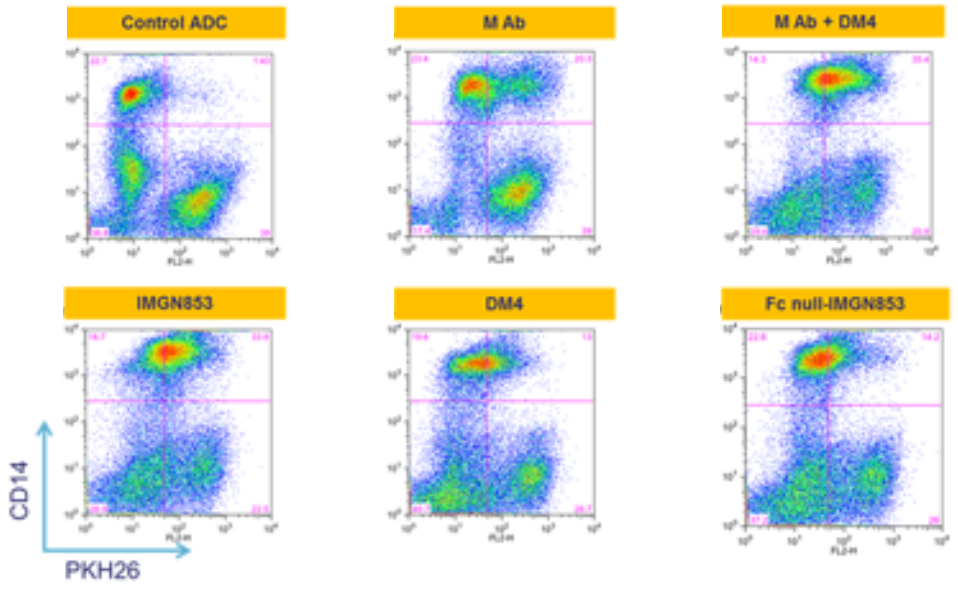
Control ADC
M Ab
M Ab + DM4
IMGN853
DM4
Fc null-IMGN853
CD14
PKH26

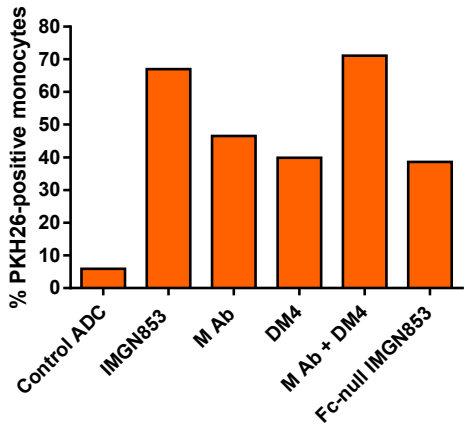
% PKH26-positive monocytes
Control ADC
IMGN853
M Ab
DM4
M Ab + DM4
Fc-null IMGN853

IMGN853 Induces Immunogenic Cell Death of KB Cells

KB cells were treated with 10 nM IMGN853 for 48 hours and immunogenic cell death markers were measured by flow cytometry (calreticulin) or ELISA (ATP and HMGB1). CRT (calreticulin), ATP, and HMGB1 (high mobility group box 1) engage CD91, P2RX7 receptors, and TLR4, respectively, and induce dendritic cells to engulf dying cells, IL1-b production and cross-presentation of tumor antigens.

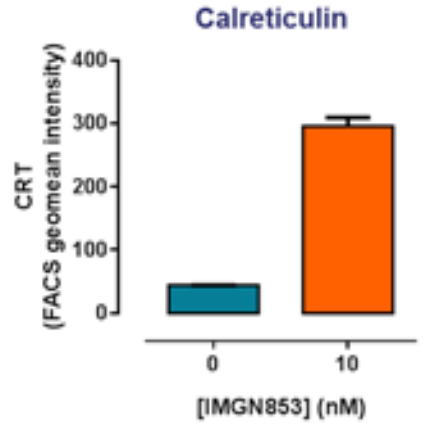
Calreticulin
CRT (FACS geomean intensity)
[IMGN853] (nM)

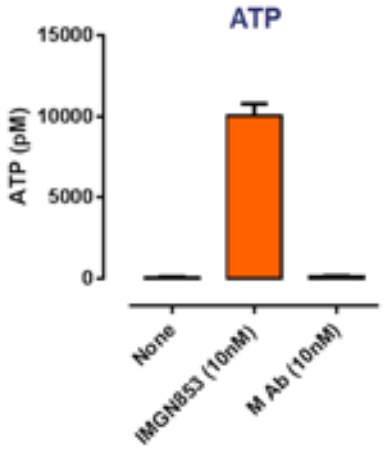
ATP
ATP (pM)
None
IMGN853 (10nM)
M Ab (10nM)

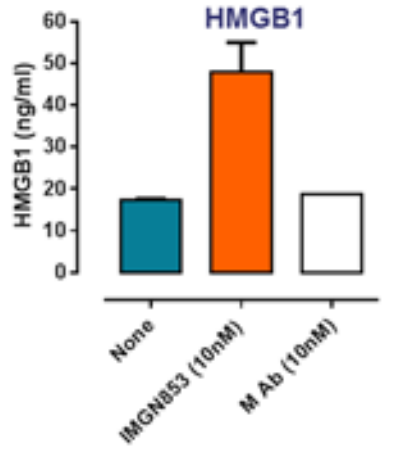
HMGB1
HMGB1 (ng/ml)
None
IMGN853 (10nM)
M Ab (10nM)

CONCLUSIONS

- **Treatment of FRα-expressing KB cells with IMGN853 induces activation of co-cultured monocytes through Fc/FcγR interaction and upregulation of immunogenic cell death markers.**
- **The activation of monocytes in the presence of tumor neo antigen would trigger T cell responses that would be further enhanced by an immune checkpoint inhibitor.**

SITC Annual Meeting 2016, 9-13 November 2016
©2016 ImmunoGen, Inc., Waltham MA, USA