ASCO 2008

Abstract#3066

The pharmacokinetics and pharmacodynamics of IMGN242 (huC242-DM4) in patients with CanAg-expressing solid tumors

A Qin[1], J Watermill[1], RA Mastico[1], RJ Lutz[1], J O'Keeffe[1], S Zildjian[1], A Mita[2], A Phan[3], and A Tolcher[4]

ImmunoGen, Inc., Waltham, MA[1]; Cancer Therapy Research Center, Institute for Drug Development, San Antonio, TX[2]; MD Anderson Cancer Center, Houston, TX[3]; and South Texas Accelerated Research Therapeutics, San Antonio, TX[4]

Abstract

Background: IMGN242 is a novel, targeted anticancer agent in development for the treatment of CanAg-expressing tumors. The compound is made by conjugating the potent cytotoxic maytansinoid, DM4, to the monoclonal antibody, huC242. In a Phase I clinical study, patients with CanAg-expressing solid tumors were treated with IMGN242 at doses ranging from 18 to 297 mg/m². The maximum tolerated dose was determined to be 168 mg/m², with ocular changes the primary dose-limiting toxicity. A Phase II study has been initiated to evaluate IMGN242 for the treatment of CanAg-expressing gastric cancer, which is highly responsive to IMGN242 in preclinical xenograft models.

Methods: The pharmacokinetics and safety of IMGN242 are being evaluated in on-going phase I and phase II studies of IMGN242 given as a single IV infusion every three weeks. Blood plasma samples were collected throughout the treatment period to determine the pharmacokinetic properties of IMGN242, to evaluate the levels of circulating CanAg and to assess the formation of human anti-IMGN242 antibodies. Assessment of CanAg expression by immunohistochemical staining was performed on tumor biopsies for all patients.

Results: Forty-five patients have been treated with IMGN242 at 8 different dose levels (18 to 297 mg/m²) in two clinical trials. Dose limiting toxicities (DLTs) included decreased visual acuity, corneal deposits and keratitis, which appeared to improve in patients where follow-up data is available. A two-phase pharmacokinetic profile was observed for IMGN242 in plasma from patients with low circulating CanAg levels (<1000 U/mL), with an initial rapid distribution phase that lasted about 48 hours, followed by a slower terminal elimination phase. Preliminary pharmacokinetic analysis reveals an elimination phase half-life for IMGN242 of about 5 days for patients with low circulating CanAg. The determined half-life in patients is similar to that predicted for IMGN242 from preclinical pharmacokinetic studies (t1/2 about 5 days in mice and 4 days in cynomolgus monkeys). Eleven patients were noted to have circulating CanAg levels greater than 1000 U/mL, although there appeared to be no correlation between high plasma CanAg and the pattern of tumor CanAg expression. High plasma CanAg levels appeared to have a marked impact on the pharmacokinetics of IMGN242 with clearance increased 3 to 5-fold in patients with high CanAg (≥1000 U/mL) compared to patients with low levels (<1000 U/mL). Cmax increased proportionally with increasing dose and was not significantly affected by circulating CanAg levels. It appeared that patients who developed study drug-related ocular toxicities had low plasma CanAg levels which may correlate with higher IMGN242 exposure in these patients.

Conclusions: The circulating CanAg level does not correlate with the tumor CanAg antigen expression in patients. The data is suggestive of a correlation or at least a trend between the level of plasma CanAg, IMGN242 exposure and the observed ocular toxicities in patients. In patients with low plasma CanAg levels (< 1000 U/ml), the dose of 168 mg/m² appeared to be associated with a notable incidence of possible study drug-related ocular toxicities (3 out of 17 in Study 101 and 3 out of 6 in Study 102). Evaluation of preclinical and clinical pharmacokinetic data indicate that the dose of 126 mg/m² in patients with low plasma CanAg levels (< 1000 U/ml) in general are likely to result in drug plasma exposures that could be efficacious. Based on the safety and thorough clinical PK/PD analyses, the Phase II study 102 was amended to treat patients with low plasma CanAg levels at the dose of 126 mg/m² and patients with high plasma CanAg levels at 168 mg/m².

Background

In the Phase I clinical study 101, patients with CanAg-expressing solid tumors were treated with IMGN242 at doses ranging from 18 to 297 mg/m². The maximum tolerated dose was determined to be 168 mg/m², with ocular changes the primary dose-limiting toxicity. Possible study drug-related ocular toxicities were also observed in 3 patients treated in the Phase II study 102 in patients with CanAg-positive gastric or gastroesophageal (GE) junction cancer. Therefore, analysis was performed to examine the relationship of pharmacokinetics and pharmacodynamics with regards to dose, plasma CanAg level, tumor CanAg antigen expression, and possible study drug-related ocular toxicities using data from both the 101 and 102 studies.

Chemical representation of structure of IMGN242

(on average, n=3.5)

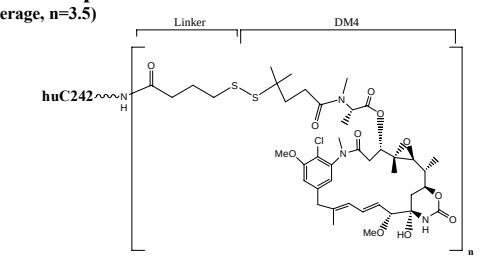
Linker
DM4
huC242
n

Objectives

Phase I Study 101:

Primary

- To determine the dose limiting toxicities (DLTs) and maximum tolerated dose (MTD) of IMGN242 administered as a single intravenous (IV) infusion once every three weeks.

Secondary

- To determine the qualitative and quantitative toxicities of IMGN242 given as a single IV infusion once every three weeks.
- To characterize the pharmacokinetics (PK) of IMGN242 given as a single IV infusion once every three weeks.
- To describe any anti-tumor activity of IMGN242 in the study population.

Phase II Study 102:

Primary

- To assess the response rate (%) of IMGN242 in patients with metastatic or locally-advanced gastric or GE junction cancer when administered as a single IV infusion once every three weeks at a defined dose (168 mg/m²).

Secondary

- To assess the duration of any responses.
- To assess progression-free survival.
- To evaluate safety and tolerability of IMGN242 administered with this schedule in the study population.
- To assess the PK of IMGN242 in this patient population.
- To assess the effect of IMGN242 on tumor uptake of [18]FDG.

Results

Baseline Demographics and Disease Characteristics of Patients in Study 101 (n=38*) and Study 102 (n=6)

	Study 101	Study 102
Median Age (years)	61.5	53
ECOG performance status of 0 or 1:		
•0	10	1
•1	26	5
Male, n (%)	28 (73.7%)	3 (50%)
Female, n (%)	10 (26.3%)	3 (50%)
Caucasian, n (%)	29 (76.3%)	6 (100%)
Asian/Pacific Rim, n (%)	1 (2.6%)	0
Hispanic, n (%)	8 (21.1%)	0
Prior Chemotherapy:		
•1 prior regimen, n (%)	1 (2.6%)	2 (33.3%)
•2 prior regimen, n (%)	5 (13.2%)	1 (16.7%)
•3 prior regimen, n (%)	3 (7.9%)	0
•≥ 4 prior regimens, n (%)	29 (76.3%)	3 (50%)
Prior Radiation Therapy:		
•1 prior radiotherapy, n (%)	6 (15.8%)	2 (33.3%)
•2 prior radiotherapy, n (%)	7 (18.4%)	1 (16.7%)
Tumor Type:		
•Colon CA, n (%)	21 (55.3%)	0
•Pancreatic CA, n (%)	2 (5.3%)	0
•Other CA, n (%)	15 (39.5%)	6 (100.0%), gastric

*Note: Data for one additional patient is not yet in the study database.

Major Toxicities in Studies 101 and 102

Phase I Study 101 (39 patients treated):

- DLTs were ocular toxicities – including corneal deposits, keratitis, and/or decreased visual acuity/blurry vision – in 2 patients treated at 223 mg/m² and 1 patient treated at 168 mg/m² who was receiving preservative-free, lubricating eye drops during study.
- Two additional patients at 168 mg/m² had mild ocular toxicities including dry eye and blepharitis that were not considered to be DLTs by the study investigators.
- At 168 mg/m², other SAEs considered to be possibly drug related were grade 3 diarrhea, reported in 1 patient who had acute renal failure/insufficiency with elevated creatinine, and 1 patient with generalized weakness, pedal edema, and inability to void for 2 days.
- All the adverse events markedly improved or returned to baseline after drug discontinuation.

Phase II Study 102 (6 patients treated):

- 1 patient developed dry eye with cornea deposits in Cycle 1 that was considered to be study-drug related.
- 1 patient had dry eye syndrome with cornea erosions in Cycle 1.
- 1 patient developed blurry vision/visual acuity decrease in Cycle 2.
- 2 patients who entered the study with liver metastases had mild-to-moderate and reversible elevations of hepatic transaminases.

Pharmacokinetic Profiles of Patients Treated with High Doses of IMGN242 in Study 101

IMGN242 concentration and circulating CanAg levels in plasma were determined by specific ELISA methods.

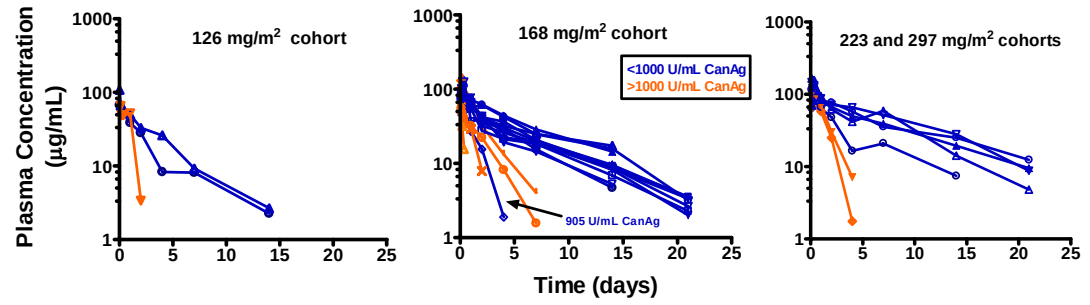
Plasma Concentration (µg/mL)
126 mg/m² cohort
168 mg/m² cohort
223 and 297 mg/m² cohorts
<1000 U/mL CanAg
>1000 U/mL CanAg
905 U/mL CanAg
Time (days)

High (>1000 U/mL) levels of CanAg in the plasma impact the PK profile of IMGN242

Relationship between Dose and Plasma CanAg Levels and IMGN242 Exposure (Cmax and total AUC)

Pharmacokinetic analysis of data from Study 101 was conducted using Pharsight WinNonlin ver 5.1.

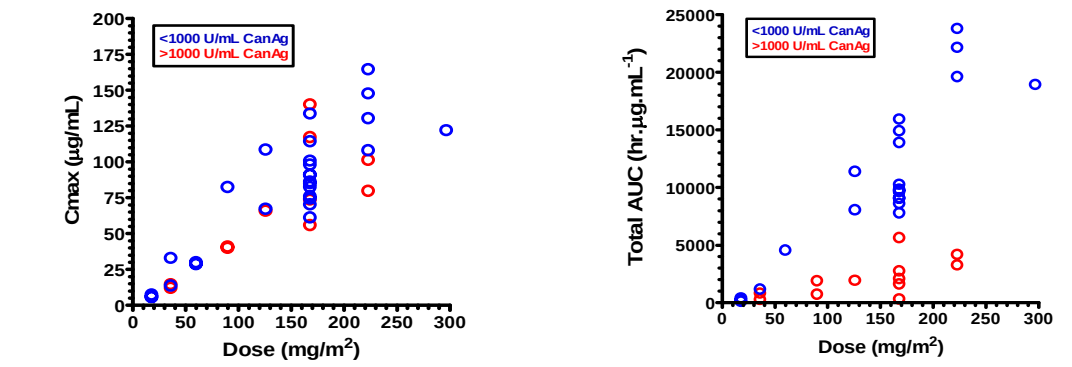
Cmax (µg/mL)
Total AUC (hr.µg.mL⁻¹)
<1000 U/mL CanAg
>1000 U/mL CanAg
Dose (mg/m²)

Impact of dose: IMGN242 exposure increased proportionally with dose. Impact of plasma CanAg levels: High (>1000 U/mL) plasma CanAg levels did not significantly impact C_{max}, but substantially lowered AUC.

Pharmacokinetic Parameters and Plasma CanAg Levels of Patients with Ocular Toxicity

Data from Studies 101 and 102

Dose mg/kg (mg/m²)	# Patients at Dose Level*	Plasma CanAg level (U/mL)	C_{MAX} µg.mL⁻¹ (Range)	$t_{½}$ h (Range)	CL mL.h⁻¹.kg⁻¹ (Range)	$AUC_{0-\infty}$ h.µg.mL⁻¹ (Range)	V_{SS} mL.kg⁻¹ (Range)
4.5 (168)	6	61.6 (2.3 -264.5)	113.7 (84.9 -154.2)	118.3 (64.3-155.3)	0.31 (0.14-0.50)	19003.4 (9068.7-33132.1)	46.2 (14.4-83.0)
6.0 (223)	2	297.3 (19.6-538.6)	139.0 (130.3-147.6)	168.5 (151.9-185.1)	0.26 (0.25-0.27)	14010.6 (13493.8-14526.7)	61.1 (52.1-69.9)

*Pharmacokinetic parameters reported as mean (range).

Relationship between Reported Ocular Toxicity, Plasma CanAg Levels, and IMGN242 Exposure

Data from Studies 101 and 102; Patients in green had ocular toxicities considered possibly or probably drug related

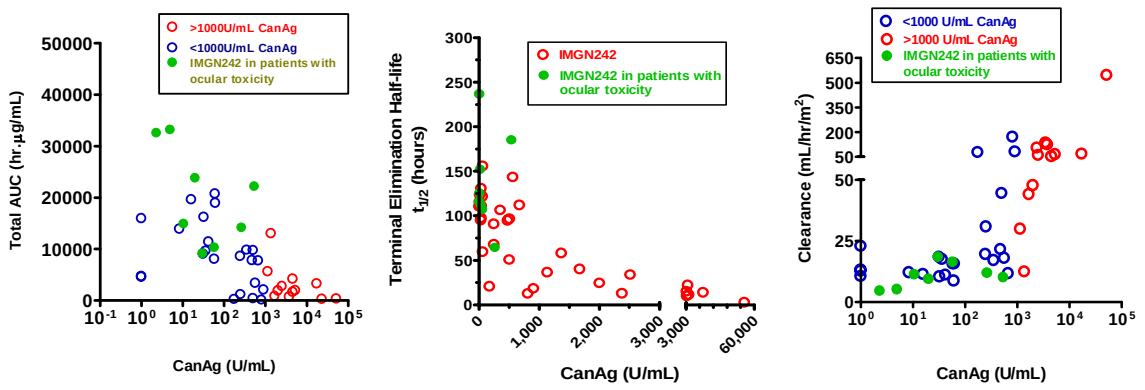
Total AUC (hr.µg/mL)
>1000U/mL CanAg
<1000U/mL CanAg
IMGN242 in patients with ocular toxicity
CanAg (U/mL)
Terminal Elimination Half-life t1/2 (hours)
IMGN242
IMGN242 in patients with ocular toxicity
CanAg (U/mL)
Clearance (mL/hr/m²)
<1000 U/mL CanAg
>1000 U/mL CanAg
IMGN242 in patients with ocular toxicity
CanAg (U/mL)

All of the patients with reported ocular toxicities had low plasma CanAg levels with relatively high AUC values, long $t_{1/2}$, and slow clearance.

Rationale for Recommended Dose for Study 102 in Gastric and GE Junction Cancers.

In vivo **efficacy of IMGN242 against human gastric tumor xenografts in SCID mice**

Cell line (ATCC #) No. mice (day of treatment)	Dose mg protein/ kg	Log_{10} cell kill (No. mice)	Complete Response No. mice	Tumor Free Survival No. mice	Comments
Gastric SNU-16 (CRL-5974) N = 5 (day 23)	0		0/5	0/5	
	1.8	0.47 (n=5)	0/5	0/5	Inactive
	3.8	1.08 (n=5)	1/5	0/5	Active
	8.6	-	3/5	1/5	Highly active
	19.7	-	5/5	3/5	Highly active
Gastric NCI-N87 (CRL-5822) N = 6 (day 5)	0		0/6	0/6	
	1.8	1.27 (n=6)	0/6	0/6	Active
	3.5	2.08 (n=6)	0/6	0/6	Highly Active
	8.3	2.93 (n=6)	1/6	0/6	Highly active
	18.9	-	5/6	0/6	Highly active

Comparison of PK Parameters between Pharmacologically Active Doses of IMGN242 in Mice and Patients Receiving 126 mg/m² in Study 101

	Dose mg/kg (mg/m²)	C_{MAX} µg.mL⁻¹ (Range)	$t_{½}$ h (Range)	CL mL.h⁻¹.kg⁻¹ (Range)	$AUC_{0-\infty}$ h.µg.mL⁻¹ (Range)	V_{SS} mL.kg⁻¹ (Range)
Mouse PK (measured)						
IMGN242	10 (30)	184.1	120.5	0.82	12140.3	129.9
Mouse PK (simulated at active doses)						
IMGN242	3.8 (11.4)	67.0	107.8	0.86	4414.8	71.1
IMGN242	1.8 (5.4)	31.7	107.8	0.86	2091.2	71.1
Human PK (measured at 126 mg/m²; mean; n = 2; Plasma CanAg < 1000 U/mL)						
IMGN242	3.4 (126)	87.9	109.3	0.36	9675.2	55.2

Comparison of PK Profiles of Pharmacologically Active Doses in Mice vs. Patients Receiving 126 mg/m² in Study 101

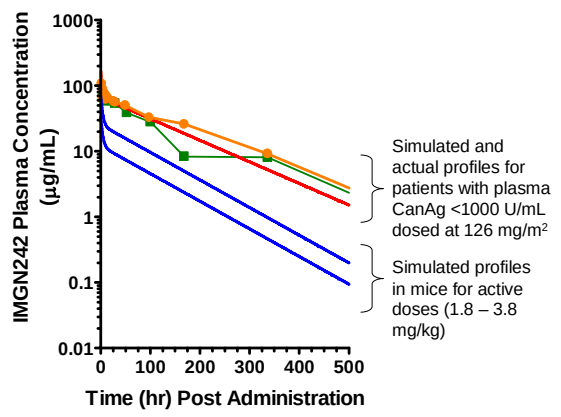
IMGN242 Plasma Concentration (µg/mL)
Time (hr) Post Administration
Simulated and actual profiles for patients with plasma CanAg <1000 U/mL dosed at 126 mg/m²
Simulated profiles in mice for active doses (1.8 – 3.8 mg/kg)

A dose of 126 mg/m² achieves, by PK analysis, a greater IMGN242 exposure in patients who have low levels of plasma CanAg than doses found to be active in mice with a single bolus IV injection

Relationship between Plasma CanAg Levels and Tumor CanAg Expression by IHC - data from Study 101 and 102.

IHC result	% of total patients (n)	% of pts with plasma CanAg >1000 U/mL (n)
Total	(44)	(12)
3 homo/hetero/focal	79 (33)	75 (9)
3 homo/hetero	68 (28)	67 (8)
3 homo	39 (17)	59 (7)
3 hetero	29 (11)	8 (1)
3 focal	11 (5)	8 (1)
2 homo, hetero, focal	9 (4)	17 (2)
1 homo, hetero, focal	2 (1)	0
Negative	7 (3)	0
N/A	7 (3)	8 (1)

There is no correlation between high levels of CanAg in the plasma and the pattern of CanAg expression on the tumor

Study 102 Case Report: Patient 0402

FDG-PET Scan Images

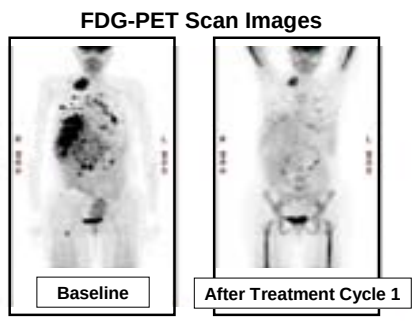
Baseline
After Treatment Cycle 1

- 46 year old female diagnosed with GE junction cancer in 2004.
- She was treated with multiple chemotherapeutic agents – including carboplatin, docetaxel and 5-FU – as well as with radiation therapy.
- Patient entered Study 102 with metastases in multiple organs including liver and lung.
- She received IMGN242 at 168 mg/m² (Cycle 1) and then at 126 mg/m² (Cycles 2 and 3); the dose reduction was due to additional changes in liver function based on laboratory tests. Patient had PD and went off study after Cycle 3.
- Her tumor had 3+ homogeneous CanAg expression by IHC. Her plasma CanAg level at study entry was 264.5 U/mL. Her IMGN242 AUC (Cycle 1) was 14123.5 h.µg.mL⁻¹

Patient had a marked biological response in Cycle 1 based on PET scans and an unconfirmed PR per RECIST in Cycle 2

Conclusions

- **High levels (>1000 U/mL) of CanAg in patient plasma reduce IMGN242 exposure (AUC) compared with low levels (<1000 U/mL) due to altered PK.**
- **Plasma CanAg levels do not correlate with the pattern of tumor CanAg expression.**
- **Ocular toxicities were observed only in patients who received high doses of IMGN242 and who also had low levels of plasma CanAg.**
- **The data support a correlation among plasma CanAg levels, IMGN242 exposure, and reports of ocular toxicities in patients.**
- **In patients with low levels of plasma CanAg, dose levels of 168 mg/m² or greater appeared to be associated with the occurrence of ocular toxicities (5 out of 24 in Study 101 and 3 out of 6 in Study 102) possibly/probably related to the study drug.**
- **Evaluation of preclinical and clinical PK data indicate that a dose level of 126 mg/m² in patients with low levels of plasma CanAg could be efficacious.**
- **A marked biological response has been observed in Study 102 in a patient with metastatic GE junction cancer. This patient had low levels of plasma CanAg.**
- **Based on safety and a thorough analyses of PK/PD data from the 101 and 102 studies to date, the protocol for study 102 was amended to have patients with low levels of plasma CanAg receive 126 mg/m² of IMGN242 and patients with high levels of plasma CanAg receive 168 mg/m² of IMGN242.**