Phase I Study of IMGN901 (huN901-DM1 or BB-10901) in Patients with Relapsed and Relapsed/Refractory CD56-Positive Multiple Myeloma.

Chanan-Khan, A.[1], Jagannath, S.[2], Munshi, N.[3], Schlossman, R.[3], Anderson, K.[3], Lee, K.[1], DePaolo, D.[1], Miller, K. C.[1], Guild, R.[4], Zildjian, S.[4], Fram, R. J.[4], Qin, A.[4]

Roswell Park Cancer Institute, Buffalo, NY[1]; St. Vincent's Comprehensive Cancer Center, New York, NY[2]; Dana Farber Cancer Institute, Boston, MA[3]; ImmunoGen, Inc., Cambridge, MA[4]

Abstract

Background: IMGN901 (huN901-DM1) is a humanized monoclonal antibody that binds with high affinity to CD56 and is covalently linked to a novel cytotoxic maytansinoid DM1. Once bound to CD56, the conjugate is internalized and releases DM1. CD56 is expressed on a variety of tumor types including small cell lung carcinoma, neuroendocrine tumors and hematological malignancies. About 70% of Multiple Myeloma (MM) patients have surface expression of CD56. Preclinical investigations demonstrated significant *in vitro* and *in vivo* anti-myeloma activity of huN901-DM1. **Objectives:** To determine the maximum tolerated dose (MTD), the dose-limiting toxicities (DLTs), and pharmacokinetics (PK) of huN901-DM1 given on a weekly schedule. **Methods:** Relapsed or relapsed/refractory MM patients who have failed at least one prior therapy and have CD56-expressing myeloma received a single IV infusion of huN901-DM1 on 2 consecutive weeks every 3 weeks. Subjects are enrolled in cohorts of 3 at each dose level. The starting dose was 40 mg/m²/week based on experience from a prior phase I trial in solid tumors. **Results:** Fourteen patients have received IMGN901, 3 patients each on 40 mg/m²/week, 60 mg/m²/week, 75 mg/m²/week, and 90 mg/m²/week cohorts, and 2 patients on the 112 mg/m²/week cohorts. No patients have experienced DLTs and no serious adverse events related to study drug were observed. In addition, no patients have experienced serious hypersensitivity reactions or evidence of HAHA or HADA formation. Our preliminary PK findings demonstrate a terminal half life of IMGN901 of about 20 hours. Immunohistochemistry performed on marrow aspirates 24 hours after IMGN901 infusion at 40 mg/m² confirmed the presence of IMGN901 on myeloma cells in the marrow. One patient treated at 60 mg/m²/week had a minimal response (MR) per European Bone Marrow Transplant criteria and had received about 15 cycles of therapy. One patient treated at 90 mg/m²/week received 4 cycles of the treatment and had a MR**.** This phase I study provides preliminary evidence of safety and clinical activity of IMGN901 in patients with CD56-positive MM who have failed established MM treatments. Targeting of IMGN901 to myeloma cells in the marrow was confirmed. Toxicity is manageable. The MTD is not yet defined and enrollment is ongoing .

Background

Figure 1. IMGN901 (Maytansinoid DM1 conjugated to huN901 antibody)

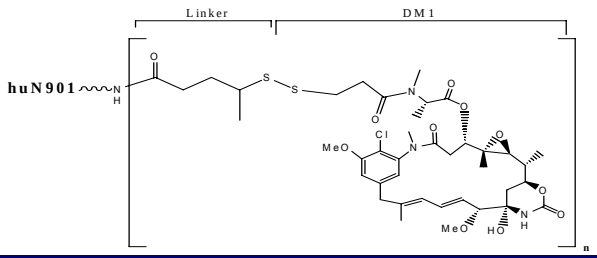
Linker
DM1
huN901
MeO
Cl
MeO
HO
n

(On average n= 3.5)

Objectives

Primary
To determine the dose-limiting toxicities (DLTs) and the maximum tolerated dose (MTD) of IMGN901 when given to patients with relapsed and relapsed/refractory CD56-positive MM by IV infusion weekly for 2 consecutive weeks every 3 weeks.

Secondary
•To determine the qualitative and quantitative toxicities of IMGN901 administered on this schedule.
•To evaluate the pharmacokinetics of IMGN901.
•To recommend a dose for Phase II clinical studies.
•To observe any evidence of anti-tumor activity with IMGN901.

Methods

Eligibility
Major Inclusion Criteria
•Patient was previously diagnosed with MM based on standard criteria.
•CD56-positive, relapsed or relapsed/refractory MM.
•Age ≥ 18 years .
•ECOG performance status ≤ 2.
•Informed consent.
•Women of child bearing potential must have a negative pregnancy test.
•Any chemotherapy or radiotherapy must be completed ≥ 4 weeks prior to Day 1.

Methods Cont'd

Major Inclusion Criteria Cont'd:
•Absolute neutrophil count (ANC) > 1000 cells/mm³, Hb ≥ 8.5 g/dL, and platelet count ≥ 75,000/mm³.
•AST (SGOT) and ALT (SGPT) ≤ 3 x upper limit of normal (ULN) and total bilirubin ≤ 1.5 x ULN.
•Creatinine ≤ 2 mg/dL.
•Left ventricular ejection fraction ≥ lower limit of normal on MUGA scan.

Major Exclusion Criteria
•Concomitant therapy with corticosteroids (except as indicated for other medical conditions or as pre-medication for blood products or study drug).
•Concomitant therapy with other antineoplastic treatments.
•Peripheral neuropathy ≥ grade 3; painful grade 2 neuropathy.
•Significant cardiac disease.
•History of multiple sclerosis or other demyelinating disease, hemorrhagic or ischemic stroke within the last 6 months, central nervous system injury with residual neurological deficit.
•Treatment with another investigational agent during the study or ≤ 4 weeks prior to Day 1.
•Previous monoclonal antibody therapy.
•Prior malignancy (within the last 3 years).

Treatment
Patients were treated in cohorts of 3 patients at increasing doses of IMGN901. IMGN901 was given as an IV infusion on two consecutive weeks every three weeks.

Trial Design
This Phase I trial is an open label, dose escalation trial in which cohorts of 3 patients receive escalating doses of IMGN901. If a DLT is observed then the cohort is expanded to six patients. MTD is defined as the highest dose at which ≤ 1 of six patients has a DLT. The dose levels to be evaluated are 40, 60, 75, and 90 mg/m². Additional dose levels will be explored if necessary.

Efficacy assessments:
Serum and urine M components are evaluated on Day 1 of each treatment cycle. Responses are assessed using European Bone Marrow Transplant (EBMT) criteria.

Pharmacokinetics and assessments of immunogenicity:
Plasma was evaluated for the presence of humoral response against the huN901 antibody component (HAHA) or against the DM1 component (HADA). Pharmacokinetic analyses were performed using standard algorithms of the non-compartmental pharmacokinetic analysis (WinNonLin 4.2, Pharsight).

Results

Table 1. Baseline Demographics and Disease Characteristics (n-13)

Median Age (years)	61.8 (49-71)
ECOG performance status	
• 0	9
• 1	2
Male, n (%)	5 (38.5%)
Female, n (%)	8 (61.5%)
Caucasian	10 (76.9%)
Black/African American	2 (15.4%)
Other: Hispanic	1 (7.7%)
Prior Chemotherapy	
• ≥ 4 prior regimens, n (%)	13 (100.0%)
Prior Radiation Therapy	
• 1 prior Radiotherapy, n (%)	2 (15.4%)
• 2 prior Radiotherapy, n (%)	1 (7.7%)
• ≥ 4 prior Radiotherapy, n (%)	1 (7.7%)

Results Continued

Table 2. Dose Escalation Scheme

IMGN901 Dose Level (mg/m²/week)	Number of Patients	Total Number of Cycles	Patients with DLT	
			First Cycle	All Cycles
40	3	4	0/3	0/4
60	3	29	0/3	0/29
75	3	7	0/3	0/7
90[a]	3	11	0/3	0/11
112[b]	2	7	0/2	0/7

Abbreviations: DLT= dose-limiting toxicity, C= Cycle
[a] Currently there is 1 active patient in the dose level.
[b] Currently there are 2 active patients.

Table 3. CTC Grade 1 and 2 Adverse Events* Considered Probably Related or Possibly Related to Study Drug (There has been no Grade 3 or 4 AE considered Probably or Possibly related to Study Drug).

System Organ Class (a) / MEDRA Term (b)	Dose Group									
	40 m/m²		60 m/m²		75 m/m²		90 m/m²	112 m/m²	Overall	
	n	(%)	n	(%)	n	(%)	n	n	n	(%)
Total Number of Patients Dosed	3		3		3		3	1	13	
Total Number (%) of Patients with Adverse Event (s)	3	100%	1	33.3%	1	33.3%	0	0	5	38.5%
Eye Disorders	1	33.3%	0		1	33.3%	0	0	2	15.4%
•Abnormal sensation in eye	1	33.3%	0		0		0	0	1	7.7%
•Ocular hyperaemia	1	33.3%	0		0		0	0	1	7.7%
•Vision blurred	0		0		1	33.3%	0	0	1	7.7%
Gastrointestinal disorders	1	33.3%	0		0		0	0	1	7.7%
•Nausea	1	33.3%	0		0		0	0	1	7.7%
General Disorders and Administration Site Conditions	0		0		1	33.3%	0	0	1	7.7%
•Asthenia	0		0		1	33.3%	0	0	1	7.7%
Infections and Infestations	1	33.3%	0		0		0	0	1	7.7%
•Nasopharyngitis	1	33.3%	0		0		0	0	1	7.7%
Investigations	0		0		1	33.3%	0	0	1	7.7%
•Alanine aminotransferase increased	0		0		1	33.3%	0	0	1	7.7%
•Asparate aminotransferase increased	0		0		1	33.3%	0	0	1	7.7%
•Blood alkaline phosphatase increased	0		0		1	33.3%	0	0	1	7.7%
Musculoskeletal and Connective Tissue Disorders	1	33.3%	0		1	33.3%	0	0	2	15.4%
•Muscle spasms	1	33.3%	0		0		0	0	1	7.7%
•Pain in extremity	0		0		1	33.3%	0	0	1	7.7%
Nervous System Disorders	0		1	33.3%	1	33.3%	0	0	2	15.4%
•Dizziness	0		1	33.3%	0		0	0	1	7.7%
•Headache	0		0		1	33.3%	0	0	1	7.7%
•Neuropathy peripheral	0		0		1	33.3%	0	0	1	7.7%

***Includes intercurrent events**
(a) Patients who had more than one event with in the same System Organ Class were counted once.
(b) Patients who had more than one event assigned to the same MedRA term were counted once.
(c) Patients who had more than one event were counted once.
(d) Percentages were based on the number of patients dosed.

Table 4. Pharmacokinetic Parameters of huN901-DM1

Parameter	Dose			
	40 mg/m²	*60 mg/m²	75 mg/m²	90 mg/m²
Cmax (µg/mL)	32.8±11.3	62.4±24.4	42.9±6.0	49.3±10.9
$t_{1/2}$ (hr)	16.7±5.2	20.5±2.6	17.1±5.1	16.5±0.8
AUC(hr.µg/mL)	791.4±406.2	1600±449.6	1011.8±49.5	1201.7±326.3
CL (mL/hr/m²)	63.9±40.3	39.8±9.7	74.2±3.7	78.3±18.8
Vss (mL/m²)	1294.6±733.8	1060.9±311.0	2106.3±163.5	1718.2±391.8

***All values are mean ± SD calculated from Cycle 1, Dose 1; exception of 60 mg/m² inclusive of cycle 5, dose 1**

Case description (Patient 202)
69 year old female patient with relapsed/refractory MM. Prior therapy included the following regimens and treatments.
•7/02 Radiation therapy (3000cGy T6-T9).
•7-11/02 VAD and thalidomide.
•11/02-2/03 Thalidomide.
•7/03-11/03 Melphalan, arsenic trioxide, and thalidomide.
•4/03-2/04 Dexamethasone.
•2/04-2/05 Lenalidomide.
•4/05 17 AAG.
•4/06 Treatment initiated with IMGN901 at 60 mg/m²/week. The patient has received 15 cycles of treatment. The patient had a minimal response characterized by a maximal reduction in serum M component of 39%, the disappearance of urine M component and no evidence of progressive disease in the skeleton or bone marrow.

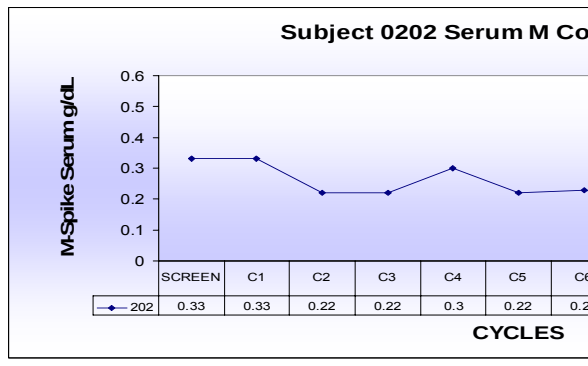
Subject 0202 Serum M Component
M-Spike Serum g/dL
0.6
0.5
0.4
0.3
0.2
0.1
0
SCREEN | C1 | C2 | C3 | C4 | C5 | C6 | C7 | C8 | 30Day FU | Off Study
202 | 0.33 | 0.33 | 0.22 | 0.22 | 0.3 | 0.22 | 0.23 | 0.2 | 0.24 | 0.51 | 0.41
202
CYCLES

Case description (Patient 212)
60 year old female patient with relapsed/refractory MM. Prior therapy included the following regimens and treatments.
•11/01-6/02 VAD and thalidomide.
•5/02-10/02 Prednisone.
•5/02-10/04 Thalidomide.
•5/03-5/03 Melphalan and arsenic trioxide.
•2/04-10/04 Lenalidomide.
•1/05-3/05; 8/06-8/06 Demethoxy.
•4/05-8/05; 6/06-6/06 Bortezomib.
•6/07 Treatment initiated with IMGN901 at 90 mg/m²/week. Patient received 4 cycles of treatment and had an MR up to date. Patient did not receive further treatment due to broken leg and rehab.

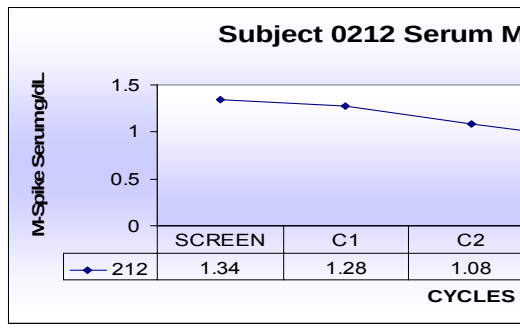
Subject 0212 Serum M Component
M-Spike Serum g/dL
1.5
1
0.5
0
SCREEN | C1 | C2 | C3 | C4
212 | 1.34 | 1.28 | 1.08 | 0.91 | 0.67
212
CYCLES

Conclusions

•This Phase I study provides evidence of safety and clinical activity of IMGN901 in patients with CD56-positive MM who have failed multiple standard therapies.
•One patient treated at 60 mg/m²/week had a minimal response (MR) per European Bone Marrow Transplant criteria and had received about 15 cycles of treatment. Another patient treated at 90 mg/m²/week had an MR up to date.
•Pharmacokinetics reveals a terminal half-life of IMGN901 of about 20 hours.
•No clinically significant myelosuppression or infusion reactions have occurred.
•No dose-limiting toxicities were observed. The MTD is not yet defined and enrollment is ongoing.