AACR-NCI-EORTC November 2009

Abstract # B237

Clinical Experience of IMGN901 (BB-10901, huN901-DM1) in Patients with Merkel Cell Carcinoma (MCC)

P.J. Woll[1], P. Lorigan[2], M. O'Brien[3], M. Villalona-Calero[4], R.A. Mastico[5], R.J, Lutz[5], C. Carrigan[5], J. O'Keeffe[5], A. Qin[5], J. O'Leary[5], and F.V. Fossella[6]

[1]Weston Park Hospital, Sheffield, UK; [2]The Christie Hospital, Manchester, UK; [3]The Royal Marsden Hospital, Sutton, UK; [4]The Ohio State University Arthur James Cancer Center & Richard J Solove Research Institute, Columbus, OH, USA; [5]ImmunoGen, Inc., Waltham, MA, USA; [6]UT MD Anderson Cancer Center, Houston, TX, USA

Abstract

Background: MCC is a neuroendocrine cancer of the skin that predominantly occurs in older individuals of northern European ancestry. While localized disease can often be treated with surgery and radiation therapy, there is a need for effective, well-tolerated chemotherapeutic options for patients with progressive or metastatic disease. Among patients with metastatic MCC, overall 5-year survival rate is 11% and median survival is in the range of 7 to 9 months. To date, no chemotherapeutic agent has been able to demonstrate a significant improvement in survival. IMGN901 is a novel CD56-targeting anticancer agent. CD56 is expressed on virtually all MCC tumors. IMGN901 consists of a potent maytansinoid cytotoxic agent, DM1, attached to a CD56-binding monoclonal antibody, huN901, using an engineered linker. Once bound to CD56, IMGN901 is internalized into the cancer cell and the DM1 is released to kill the cell via inhibition of the polymerization of tubulin. **Objective:** Study 002 is an ongoing phase I trial established to determine the maximum tolerated dose (MTD), pharmacokinetics (PK) and activity of IMGN901 in patients with CD56-expressing solid tumors, including patients with MCC. **Methods:** IMGN901 is administered intravenously as monotherapy daily for 3 consecutive days every 3 weeks. **Results:** Six MCC patients have been enrolled in Study 002 in addition to patients with other types of CD56+ solid tumors. Clinical benefit has been observed in 3 of these 6 patients including one complete response (CR), one partial response (PR) and one stable disease (SD). The patient who experienced a CR has remained disease free for more than 4 years. The patient who experienced a PR received 1 cycle of IMGN901 treatment, but had a serious adverse event and declined further treatment. This patient had sustained clinical benefit for more than 5 months, including additional tumor shrinkage after attaining a PR. The patient who experienced SD entered Study 002 with bone metastases and had received 3 prior therapies. This patient had SD lasting approximately 3 months. Overall, a clinical benefit rate (CR+PR+SD) of 50% has been observed. The 6 MCC patients received IMGN901 at doses ranging from 36 to 75 mg/m^2/day. Earlier this year, we reported pooled data from two studies in CD56+ solid tumors summarizing the encouraging experience with IMGN901 for the treatment of small-cell lung cancer (SCLC). SCLC and MCC have shared morphologic characteristics and a common clinical course.

Background

Figure 1. IMGN901 (Maytansinoid DM1 conjugated to huN901 antibody)

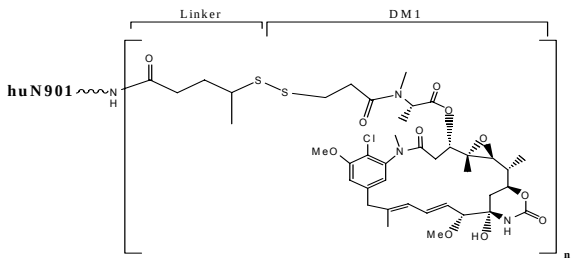
Linker
DM1
huN901
MeO
Cl
MeO
HO
n

(On average n= 3.5)

Pre-Clinical Assessment of CD56 Expression in MCC

Immunohistochemical (IHC) staining show that CD56 is expressed in all the MCC samples (n=22 [a]) examined

Table 1. Immunoreactivity of anti-CD56, clone 1B6, with formalin fixed paraffin embedded (FFPE) biopsy samples of MCC.

Slides containing FFPE sections (5 μM) of MCC were heat-induced antigen retrieved using a decloaker with citrate buffer (pH 6.0). Primary test (murine anti-huCD56 clone 1B6 IgG1, 0.75 μg/mL; Leica) and primary negative control (murine IgG1, 0.75 μg/mL; Coulter) and secondary (biotin-horse-anti-mouse, H&L affinity purified; Vector) antibodies were prepared in diluent (PBS containing 2% Horse Serum). The Avidin-Biotin-peroxidase Complex (ABC; Vector) was used as the detection system, and DAB (DAKO) was used as the substrate

Score [b]	Total # of Merkel tumor samples	# of primary tumor samples	# of metastasis samples
3 or 3+ homo	12	6	6
2-3 homo & 3 hetero	4	3	1
1-2 hetero & 1-3 focal	6	5	1

[a] The samples include 3 samples respectively from 3 patients in Study 002. The remaining MCC tissue samples were acquired from Tissue Solutions, Proteogenex, and the Cooperative Human Tissue Network.

[b] The IHC staining of CD56 is scored based on the following criteria:

(1) Scoring was based specifically on membrane staining of tumor cells

(1) Intensity of staining is ranked as follows:

0=negative, 1=weak, 2=moderate, 3=strong, 3+=very strong

(2) Uniformity of staining is ranked by % of tumor cells which stain in sample:

homogeneous (homo): >75%, heterogeneous (hetero): 25-75%, focal: <25%

Objectives of Study 002

Primary

- To determine the safety, tolerability and MTD of IMGN901

Secondary

- To determine the pharmacokinetics (PK) of IMGN901
- Preliminary assessment of the efficacy of IMGN901 when administered daily for 3 days to patients with relapsed or refractory SCLC, other pulmonary tumors of neuroendocrine origin, non-pulmonary small cell carcinomas, metastatic carcinoid tumors or other CD56+ solid tumors

Study 002 Methods

Major Inclusion Criteria

- Histologically/cytologically proven SCLC or other CD56+ solid tumors
- Tumors other than SCLC require confirmation of CD56 expression prior to enrollment
- With the exception of carcinoid and neuroendocrine tumors (which may not have been treated with chemotherapy previously), patients must have received at least 1 but ≤ 3 prior chemotherapy regimens
- Measurable disease
- ECOG performance status of 0-2
- Absolute neutrophils ≥ 1.5 x 10^9/L, Hb ≥10 gm/dL and platelets ≥ 100 x 10^9/L
- Cr ≤ 1.5 x upper limit of normal (ULN); AST and ALT ≤ 2.5 x ULN and bilirubin ≤ 3 x ULN
- Pancreatic function, amylase and lipase (when available at the site), within normal limits

Major Exclusion Criteria

- Chemotherapy, other investigational agents or radiotherapy within 4 weeks prior to study entry
- Known CNS metastases
- History of multiple sclerosis or other demyelinating disease, Eaton-Lambert syndrome
- History of pancreatitis
- Serious infection or other concomitant illness that would significantly interfere with the study outcome

Summary of Safety Results in Study 002

- A total of 52 patients with SCLC, neuroendocrine/carcinoid tumors, and other CD56-positive solid tumors have received IMGN901 at doses from 4 to 94 mg/m^2/day. Among them, 6 patients had MCC
- In general, IMGN901 is well-tolerated, and not associated with grade 3 / 4 myelosuppression
- Headache, nausea, fatigue, vomiting and peripheral sensory neuropathy were the most commonly reported adverse events related to IMGN901
- Dose-limiting headache and aseptic meningitis-like symptoms were reported at the dose of 75 mg/m^2/day. With prophylactic measures (a slowed infusion rate and steroid administration prior to IMGN901 infusion), headache and meningitis-like symptoms were not observed in additional patients treated at 75 mg/m^2/day
- Other dose-limiting toxicities (DLTs) reported on the study were grade 4 fatigue (1 pt at 16 mg/m^2/day), grade 3 peripheral sensory neuropathy/paraesthesia (1 pt each at 48 and 75 mg/m^2/day), grade 3 headache and pain in back and shoulder (1 pt at 94 mg/m^2/day), and grade 3 myalgia (1 pt at 94 mg/m^2/day)
- One MCC patient treated at 60 mg/m^2/day developed reversible posterior leukoencephalopathy syndrome (RPLS) after receiving 1 cycle of IMGN901 treatment. The patient had confusion, agitation, impaired memory and vision with MRI findings suggestive of RPLS.
- As 2 of 2 patients treated at 94 mg/m^2/day developed DLTs, 75 mg/m^2/day has been defined as the MTD.
- The study is ongoing with continued patient enrollment at the dose of 75 mg/m^2/day to further define the activity and tolerability of IMGN901 when dosed at its MTD.

IMGN901 Activity in SCLC Observed in Two Studies

Combined Experience in SCLC Patients (n = 68) from Study 002 and Study 001*

Objective Responses

One confirmed PR in second-line setting without evidence of progressive disease (PD) for 169 days (24 weeks)

One unconfirmed PR as third-line treatment without evidence of progressive disease for 58 days (8.3 weeks).

Stable Disease

Fifteen patients who received IMGN901 as ≥ 2nd line therapy had estimated TTP ranging from 77 to 168 days. This compares favorably to median TTP of 13.3 weeks (93 days) seen with topotecan as 2nd line treatment for SCLC (von Pawel *et al*., 2009, *JCO*, 17: 658-667).

Of these 15 patients, 8 patients received IMGN901 as ≥ 3rd-line treatment with estimated TTP ranging from 77 to 138 days.

Estimated Clinical Benefit Rate (CBR) = 25% (17 of 68 patients)

CBR = PR + SD (defined as non-progression for at least 77 days)

Represents second and ≥ third-line patient populations.

*Note: Study 001 was another clinical study of IMGN901 in CD56-positive solid tumors but with a different dosing schedule (weekly x 4 every 6 weeks).

Summary of Clinical Results in MCC Patients

Table 2. Clinical results in MCC patients

	Prior Treatment			Status at Study Entry			IMGN901 Therapy				
Pt #	Prior Chemotherapy and Response	Time on most recent (days)	XRT	Site(s) of Metastases	Age	Tumor CD56 Staining[a]	Dose Received (mg/m^2/day)	# Cycles Received	Reason Off Study	Time from 1st Dose to PD	Best Response
406	**1)** Etoposide /Cisplatinum (VP/CDDP) x 6 → PD	90	Cheek	Left forearm, local recurrence	56	3 homo	36	6	Completed Study	Not had PD to date. Has been disease free for ~ 4 yrs.	CR
417	**1)** VP/Carboplatin (VP/Carbo) x 6 → PD	103		Multiple lower extremity and foot lesions	68	+	60	1	AE (RPLS)	Not had PD to date. Has been 5 mo (as of 8/09) since last dose of IMGN901.	PR
505	**1)** VP/Carbo x 4 → CR **2)** Doxorubicin (Doxo) x 1→ CR **3)** Doxo/Vincristine (VCR) / Cyclophosphamide (Cyclo) x 8 → PR	159	Scalp, lymph node	Bone	60	3 homo	75	4	PD	79 days	SD
410	**1)** VP/Carbo x 4 → CR	70		Right forearm, right axilla	78	+	48	1	PD	13 days	PD
211	**1)** VP/Carbo x 4 → PD **2)** Doxo/Cyclo x 2 → PR	108	Spine	Left axilla node, left sub-pectoral mass, rib mass	69	+	60	2	PD	30 days (clinical PD)	PD
507	**1)** VP/Carbo x 6 → CR **2)** Doxo/VCR/Cyclo x 6 → PD **3)** Irinotecan/ Carbo x 2→ PD **4)** VP alone x 1→ PD	34	Head, neck	Liver, peritoneum, retro-peritoneal adenopathy, mediastinal adenopathy	62	3 homo	75	1	PD	21 days	PD

[a)] "+" is the score based on previous path report.

Table 3. Pharmacokinetic Parameters of IMGN901 in MCC Patients versus the Mean Pharmacokinetic Parameters of IMGN901 in Patients with other CD56+ Tumors within the Same Dose Group

Plasma concentrations of IMGN901 were determined using validated ELISA-based methods. Pharmacokinetics analyses were performed using standard algorithms of the non-compartmental pharmacokinetic analysis (WinNonLin 5.2.1, Pharsight).

Dose mg/m^2	Patient (Best Response)	Cmax ug/mL	$t_{1/2}$ hr	AUC0-∞ hr*ug/mL	CL mL/hr/m^2	Vss mL/m^2
36	406 (CR)	47.8	19.9	847	42.5	808
	Other CD56+ Tumors, N=3	32.4 ±8.9	17.1 ±1.9	711 ±166	52.7 ±13.6	1190 ±359
48	410 (PD)	NA				
	Other CD56+ Tumors, N=5	69.4 ±34.3	20.3 ±3.7	1323 ±345	38.9 ±12.5	898 ±305
60	211 (PD)	NA				
	417 (PR)	54.4	25.4	2036	29.5	1062
	Other CD56+ Tumors, N=4	69.9 ±28.1	26.8 ±12.9	3066 ±2218	33.6 ±27.8	823 ±433
75	505 (SD)	37.7	36.4	1778	42.2	2153
	507 (PD)	63.7	29.9	2949	25.4	982
	Other CD56+Tumors, N=7	92.7 ±40.3	27.9 ±8.2	4082 ±2731	24.8 ±14.2	822 ±402

*N indicates the number of patients measured for pharmacokinetic parameters and all parameters are expressed as Mean ± SD

** NA: not available.

Case Reports

Case Description (Patient 406)

- October 2003: 55-year old female diagnosed with MCC of the right cheek
- January 2004: wide excision, neck dissection and reconstruction, tumor close to margin and 5/21 lymph nodes positive
- February to March 2004: Adjuvant RT (46Gy)
- May to August 2004: chemotherapy (6 cycles cisplatin/etoposide)
- September 2004: restaging CT scan showed no disease
- November 2004: new tumor nodules in face and neck excised
- December 2004: left forearm nodule biopsy revealed CD56+ MCC
- January 2005: disease in right cheek and left forearm
- January 2005: Began therapy with IMGN901 at 36 mg/m^2/day x 3 days every 21 days - had PR at Cycle 1 and CR at Cycle 3. Patient received 6 cycles of IMGN901 treatment. Currently remains in clinical remission for more than 4 years.

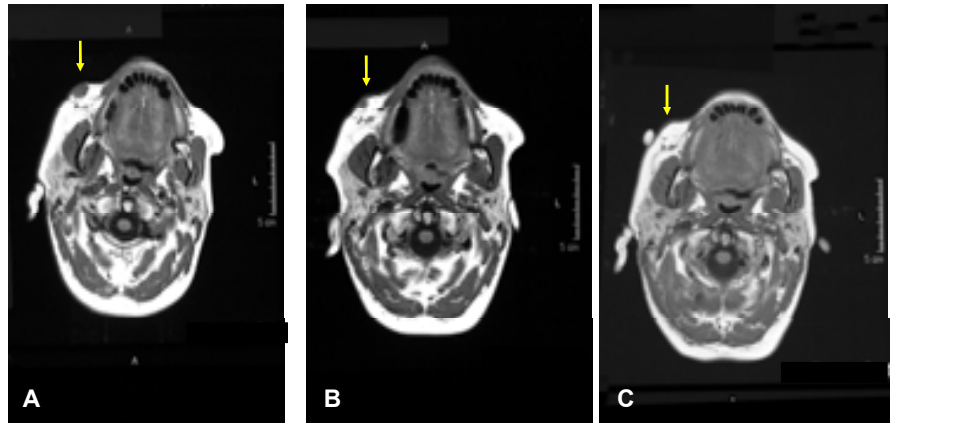
A
B
C

(A) Baseline: Jan-06-05 Rt. Cheek Lesion measures 13x9 mm

(B) Post 1 Cycle: Jan-28-05 Lesion measures 9x7 mm

(C) Post 4 Cycle: Apr-22-05 Lesion measures 0x0 mm

Case Description (Patient 417)

- 68-year old female diagnosed with MCC, presented with multiple cutaneous lesions
- August to December 2008: Received chemotherapy (6 cycles carboplatin/etoposide)
- Developed PD within 1 month after stopping chemotherapy
- February 2009: Treated with IMGN901 at 60 mg/m^2/day for 1 cycle
- IMGN901 discontinued after 1 cycle – patient developed RPLS
- June 2009: PR confirmed by CT scan approximately 9 weeks after initial CT scan documenting PR
- Continued to show improvement in skin lesions at 8 months based on clinical exam

February 2009 Prior to IMGN901

August 2009 After IMGN901 (1 cycle)

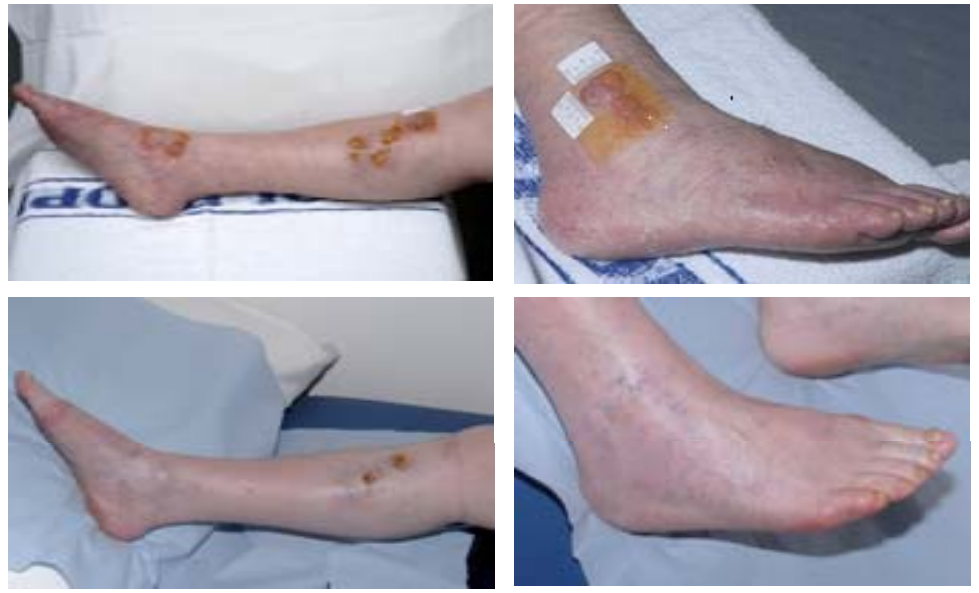

Conclusion

- Among the small group (n=6) of patients with relapsed, metastatic MCC who have been treated with IMGN901, highly encouraging activity has been observed. Of particular note is a CR that has been sustained for at least 4 years and a PR that has been ongoing for several months.
- These findings are supported by encouraging findings with IMGN901 when used for the treatment of SCLC (data reported at the 2009 World Conference on Lung Cancer), a cancer that has similarities to MCC in morphology and clinical course.
- IMGN901 has shown an encouraging safety and tolerability profile across three clinical studies in which it has been investigated.
- MCC patients have IMGN901 plasma exposures comparable to those seen in other CD56+ tumors at the same dose.
- These findings support further investigation of IMGN901 for the treatment of CD56+ solid tumors including MCC.