ASCO June 2010

Abstract # 3058

A Phase I Dose-Escalation Study of IMGN388 in Patients with Solid Tumors

D. Thompson[1], A. Patnaik[2], J. Bendell[1], K. Papadopoulos[2], J. Infante[1], C. Carrigan[3], V. Vyas[3], R.A. Mastico[3], D. Johnson[3], A. Qin[3], J. O'Leary[3], and A.W. Tolcher[2]
Sarah Cannon Research Institute, Nashville, TN[1]; South Texas Accelerated Research Therapeutics, San Antonio, TX[2]; ImmunoGen, Inc., Waltham, MA[3]

IMMUNOGEN, INC.

Background

- IMGN388 is a novel antibody-drug conjugate (ADC) composed of an integrin-targeting fully human antibody with the maytansinoid, DM4, attached via a covalent bond.
- Its target is an α_v integrin and is expressed in a wide variety of solid tumors and also on endothelial cells in the process of forming new blood vessels.
- In preclinical testing, IMGN388 has shown both anti-angiogenic activity and direct cytotoxic effect on tumor cells with pronounced activity against human xenograft lung, colon, pancreatic, ovarian and breast tumors in nude rat models.
- Study 0201 is a first-in-human, dose-escalation Phase I trial with IMGN388.

Structure of IMGN388

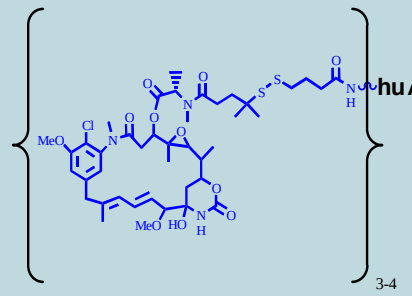
MeO
Cl
N
O
O
N
O
S
S
N
H
hu Ab
MeO
HO
H
3-4

Mechanism of Action

•Bind to targets on the surface of cancer cells

•Brought into the cell by natural processes

•Once inside, the maytansinoid agent is freed and able to kill the cancer cell

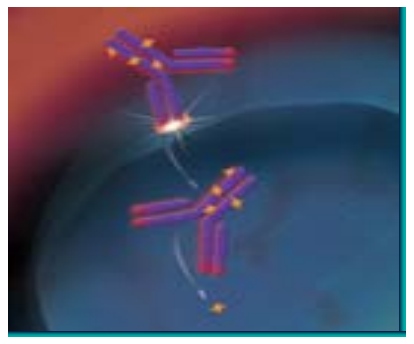

Anti-Tumor Activity of IMGN388 *in Vivo**

Nude rats bearing established subcutaneous human tumor xenografts were treated with IMGN388 as repeated IV bolus injections of 0.5 to 15 mg/kg (1 mg conjugate protein/kg, ~5.2 mg/m² in rat, equivalent to 0.015 mg linked DM4/kg).

A549, Human Lung Carcinoma

MDA-MB-231.OT.F2, Human Breast Carcinoma

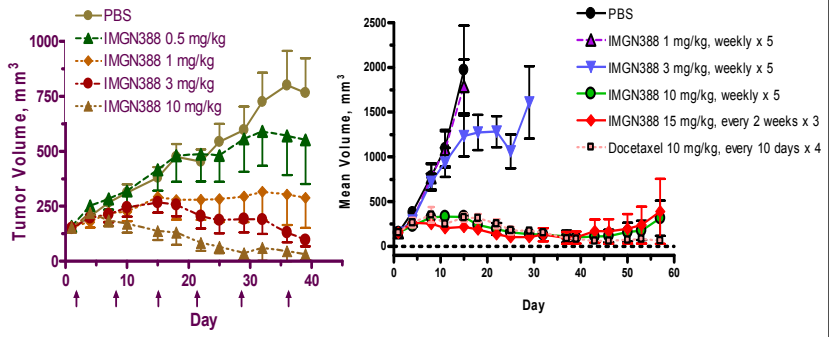
PBS
IMGN388 0.5 mg/kg
IMGN388 1 mg/kg
IMGN388 3 mg/kg
IMGN388 10 mg/kg
Tumor Volume, mm³
Day
PBS
IMGN388 1 mg/kg, weekly x 5
IMGN388 3 mg/kg, weekly x 5
IMGN388 10 mg/kg, weekly x 5
IMGN388 15 mg/kg, every 2 weeks x 3
Docetaxel 10 mg/kg, every 10 days x 4
Mean Volume, mm³
Day

* C.A. Vater, C. Manning, H. Millar, F McCabe, Q. Chen, G.M. Anderson, R. Steeves, K. Lai, S. Connaughton, V. Vyas, M. Mata, U. Prabhakar and R.J. Lutz. Anti-Tumor Efficacy of the Integrin-Targeted Immunoconjugate IMGN388 in Preclinical Studies. *AACR-NCI-EORTC 2008* Poster Presentation (Abstract #529).

Objectives of Clinical Study 0201

Primary
- To evaluate the safety and pharmacokinetics (PK) of IMGN388

Secondary
- To assess the pharmacodynamics, immunogenicity, and antitumor activity

Methods

Eligibility

Major Inclusion Criteria
- Histologically confirmed solid tumor that is metastatic or unresectable and for which standard curative or palliative measures do not exist or are not effective
- ECOG performance status of 0-2
- Standard oncology Phase I lab inclusion criteria

Major Exclusion Criteria
- Residual toxicities from previous therapy that are ≥Grade 1 (Neuropathy >Grade 1)
- History of uveitis within 6 months of first dose of study agent, or active bleeding
- Known allergies to human proteins or to therapeutic antibodies
- Standard exclusion criteria for Phase I oncology studies

Trial Design
- New cohorts of 3 patients received increasing doses of IMGN388 according to the standard oncology 3+3 design.
- Immunohistochemical (IHC) assessment of tumor specimens is not mandated prior to enrollment into the dose escalation phase, but will be required for inclusion into the expansion cohort
- Patients with any type of solid tumor are eligible for enrollment in the dose-escalation phase. In contrast, the expansion cohort will be limited to patients with melanoma or cancer of the lung, breast, or ovary with at least 2+heterogeneous expression of the antigen

Study Drug Administration
- IMGN388 is administered as an intravenous (IV) infusion once every 3 weeks

Results

Table 1. Patient Demographics and Disease Characteristics (N=32) [a]

Median Age in Years	61.5
Male	14
Female	18
Race	
• Caucasian	31
• Black/African American	1
• Hispanic/Latino	5
ECOG Performance Status at Screening	
• 0	15
• 1	16
• 2	1
Prior Chemotherapy [b]	
• None	2
• 1 Prior Regimen	3
• 2 Prior Regimens	4
• ≥ 3 Prior Regimens	22
Prior Radiotherapy [b]	
• None	18
• 1 Prior Radiotherapy	7
• ≥ 2 Prior Radiotherapy	6

[a] As of the data cut-off date of 30 April 2010, a total of 32 patients received the study drug at doses ranging from 5 to 130 mg/m².
[b] One patient's data is not available in the database yet.

ImmunoGen would like to thank the patients and their families for their participation in this trial.

Results Continued

Table 2. Cancer Types of Patients on Study 0201 (N=32)

Types	Number of Patients (%)
Ovarian Carcinoma	4 (13%)
Colorectal Cancer	4 (13%)
Neuroendocrine Carcinoma	3 (10%)
Non-Small Cell Lung Cancer (NSCLC)	3 (10%)
Prostate Cancer	3 (10%)
Breast Cancer	2 (6%)
Pancreatic Cancer	2 (6%)
Others*	11 (33%)

*One patient each with adrenal carcinoma, endometrial cancer, fibrous histiocytoma, gastric cancer, Kaposi's sarcoma, peritoneal mesothelioma, renal cell carcinoma, retroperitoneal sarcoma, small bowel adenocarcinoma, bladder and uterine cancer.

Table 3. Expression of the α_v Integrin Target by IHC Evaluation on Archived Tumor Samples from Patients Enrolled on Study 0201 (N=23*)

Target Staining	Patients n (%)
3/Homo	3 (13%)
3/Homo - Hetero	1 (4%)
3/Hetero	7 (30%)
2/Homo – 3/Hetero	1 (4%)
2/Hetero	4 (17%)
1/Hetero	2 (9%)
0/Not present	5 (22%)

•IHC assessment was not performed for all patients as it was not possible to retrieve tumors specimens for some patients.

Scoring Methods for the IHC Staining

Intensity (amount of stain)	
0	Negative
1	Weak
2	Moderate
3	Strong
3+	Very Strong

Uniformity (number of stained cells)	
0	Negative
Focal	< 25%
Heterogeneous (hetero)	25-75%
Homogeneous (homo)	>75%

● Cellular membrane staining was scored.

Figure 1. Target Staining by IHC on a Tumor Sample from a NSCLC Patient (0212)

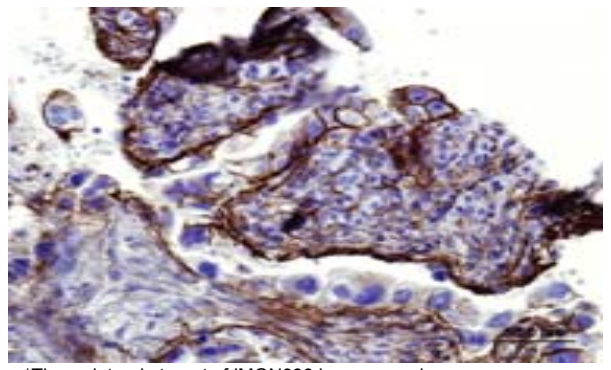

*The α_v integrin target of IMGN388 is expressed on vessels, stroma, and tumor cells

Table 4. Pharmacokinetic Parameters of IMGN388 in Study 0201
Plasma concentrations of IMGN338 were determined using validated ELISA-based methods. Analyses were performed using standard algorithms of the non-compartmental PK analysis (WinNonLin 5.2.1, Pharsight).

Parameter	Dose [b, c]				
	30 mg/m²	45 mg/m²	60 mg/m²	80 mg/m²	105 mg/m²
	N=3 [a]	N=6	N=3	N=3	N=3
Cmax (µg/mL)	12.7 ± 3.5	15.9 ± 2.3	28.1 ± 2.6	47.1 ± 4.6	62.9 ± 18.6
$t_{1/2}$ (h)	21.9 ± 5.1	25.6 ± 2.9	29.9 ± 8.6	31.6 ± 6.0	30.3 ± 7.0
AUC (h.µg/mL)	431 ± 33.6	745 ± 91.3	1527 ± 710	2287 ± 587	3135 ± 1069
CL (mL/h/m²)	70.0 ± 5.8	60.8 ± 7.5	48.1 ± 28.9	36.9 ± 11.1	36.7 ± 14.3
Vss (mL/m²)	2134 ± 670	1956 ± 161	1668 ± 625	1483 ± 367	1256 ± 195

[a] N = the number of patients measured for pharmacokinetic parameters. All parameters are expressed as Mean ± SD.
[b] PK data for the 130 mg/m² dose level are not available yet.
[c] PK samples were also used to test the presence of humoral responses against the antibody component of IMGN388 (HAHA) or against the DM4 component (HADA) and preliminary data suggest no evidence of their presence.

Figure 2. Maximal Observed Plasma Concentration and Exposure (AUC) for IMGN388

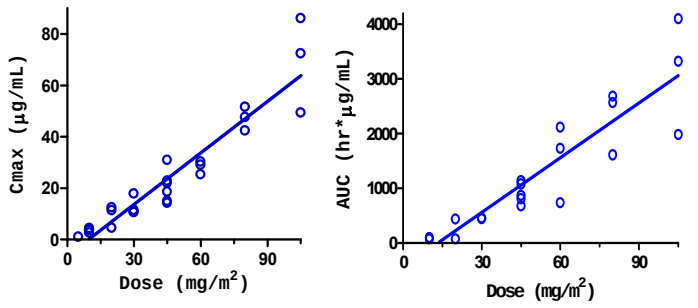
Cmax (µg/mL)
Dose (mg/m²)
AUC (hr*µg/mL)
Dose (mg/m²)

Table 5. Grade 1 or 2 Adverse Events (AEs) [a] Considered at Least Possibly Related to Study Drug

AEs [b]	Dose Levels (mg/m²)								
	5 (N=3)	10 (N=4)	20 (N=3)	30 (N=4)	45 (N=6)	60 (N=3)	80 (N=3)	105 (N=3)	130 (N=2)
Anorexia		1					1		
Nausea		2		1	1	1	2		
Vomiting				1	2		2	1	
Diarrhea		1					1		
Fatigue	1						1		
Headache					1	1	1		
Peripheral Neuropathy				1				1	
Flushing								1	
Mucosal Inflammation						1			
Thrombocytopenia					1				
Neutropenia								1	
Increased AST [c]								1	
Increased GGT [c]								1	
Increased Glucose								1	
Blood Urine							1		
Hematochezia					1				
Muscle Spasms								1	

[a] As of the data cut-off date of 30 April 2010, no reported grade 4 AE. Only 2 patients reported grade 3 AEs: 1 patient treated at 45 mg/m² had grade 3 headache and confusion; another patient treated at 80 mg/m² had grade 3 nausea and vomiting. The grade 3 headache is a dose-limiting toxicity (DLT) and it has been the only DLT on the study.
[b] Grade 1 or 2 AEs that were reported once in patients who received the study drug at 30 mg/m² or less but not at higher doses were not shown in this table.
[c] AST: aspartate aminotransferase; GGT: gamma-glutamyltransferase;

Table 6. Patients on IMGN388 Treatment with Stable Disease (SD) for 3 Cycles or Better

Patient	Dose (mg/m²)	Tumor Type	Target Expression	Lines of Prior Chemotherapy	Response	Cycles on IMGN388
0203	45	NSCLC	2/Hetero	2	SD	6
0116	60	Uterine	3/Hetero	1	SD	3
0208	80	Breast	Not available	10	SD	4
0210	80	Prostate	3/Homo	2	SD	6
0211	105	Neuroendocrine	Not available	2	SD	7*
0212	105	NSCLC	3/Hetero	5	SD	5*

*Patients still on the study as of 18 May 2010. All patients receiving the 130 mg/m² dose level also were still on study

Conclusions

- IMGN388 has been well tolerated through doses of 130 mg/m² to date. Preliminary immunogenicity data did not suggest the presence of HAHA or HADA.
- The MTD has not yet been reached and the dose escalation continues.
- One DLT of grade 3 headache was seen in the 45 mg/m² cohort. This side effect has been seen with other antibody-containing compounds and has been well-controlled with steroid prophylaxis prior to infusion. For subsequent patients steroid prophylaxis has been used with no further grade 3 headache seen.
- Maximal IMGN388 plasma concentration and exposure (AUC) generally increases linearly (data through 105 mg/m²). Elimination half-life has been consistent across dose levels.
- Archived tumor specimens have been collected for 72% of patients thus far. The α_v integrin target staining by IHC has been seen in the majority of patients, and appears to be consistently seen among patients with SD for 3 cycles or better.
- Encouraging preliminary activity has been noted.